Comprehensive Analysis of Immunoglobulin E Levels in Healthy Donors and Patients With Indolent Systemic Mastocytosis Enrolled in the PIONEER Trial of Avapritinib

Sigurd Broesby-Olsen,[1] Frank Siebenhaar,[2,3] Cem Akin,[4] Marcus Maurer,[2,3,†] Tracy I. George,[5] Hui-Min Lin,[6] Benjamin Lampson,[6] Scott Ribich,[6] Alexandra Grassian,[6] Guang Yang,[6] Rachel L. Erlich,[6] Karin Hartmann[7]

[1]Department of Dermatology and Allergy Centre, Odense University Hospital, Odense, Denmark; [2]Institute of Allergology, Charité–Universitätsmedizin Berlin, Berlin, Germany; [3]Corporate member of Freie Universität Berlin and Humboldt-Universität zu Berlin, Berlin, Germany; [4]University of Michigan; Ann Arbor, MI; [5]Huntsman Cancer Institute, ARUP Laboratories, University of Utah, Salt Lake City, UT; [6]Blueprint Medicines Corporation, Cambridge, MA; [7]Division of Allergy, Department of Dermatology, University Hospital Basel and University of Basel, Basel, Switzerland

[†]Deceased

Poster Number 521

Introduction

- Indolent systemic mastocytosis (ISM) is a clonal mast cell disease driven by the *KIT* D816V mutation in ~95% of cases[1] and patients often suffer from life-long debilitating symptoms and poor quality of life with significant comorbidity[2–5]
- Historically, the prevalence of systemic mastocytosis (SM) has been estimated at 1 in 10,000 people,[6–8] although a recent study suggests it could affect up to 1 in 5,000 people[9]
- ISM changes the composition of the innate and adaptive immune system in ways that are still being understood
 - The secreted antibody immunoglobulin E (IgE) binds to the high-affinity receptor for the Fc region of IgE (FcƐRI) on the surface of mast cells
 - Binding of antigen to the FcƐRI-IgE surface complex triggers mast cell degranulation and mediator release
 - Evidence suggests the functions of KIT and FcƐRI-IgE are closely interrelated in mast cell biology (**Figure 1**)[10]
 - Higher total IgE levels are one of the risk factors for anaphylaxis in SM, though *KIT*-mutant mast cells are also activated even with minimal or no antigen binding[11]

Figure 1. Interaction of omalizumab with IgE and avapritinib with *KIT*-mutant mast cells

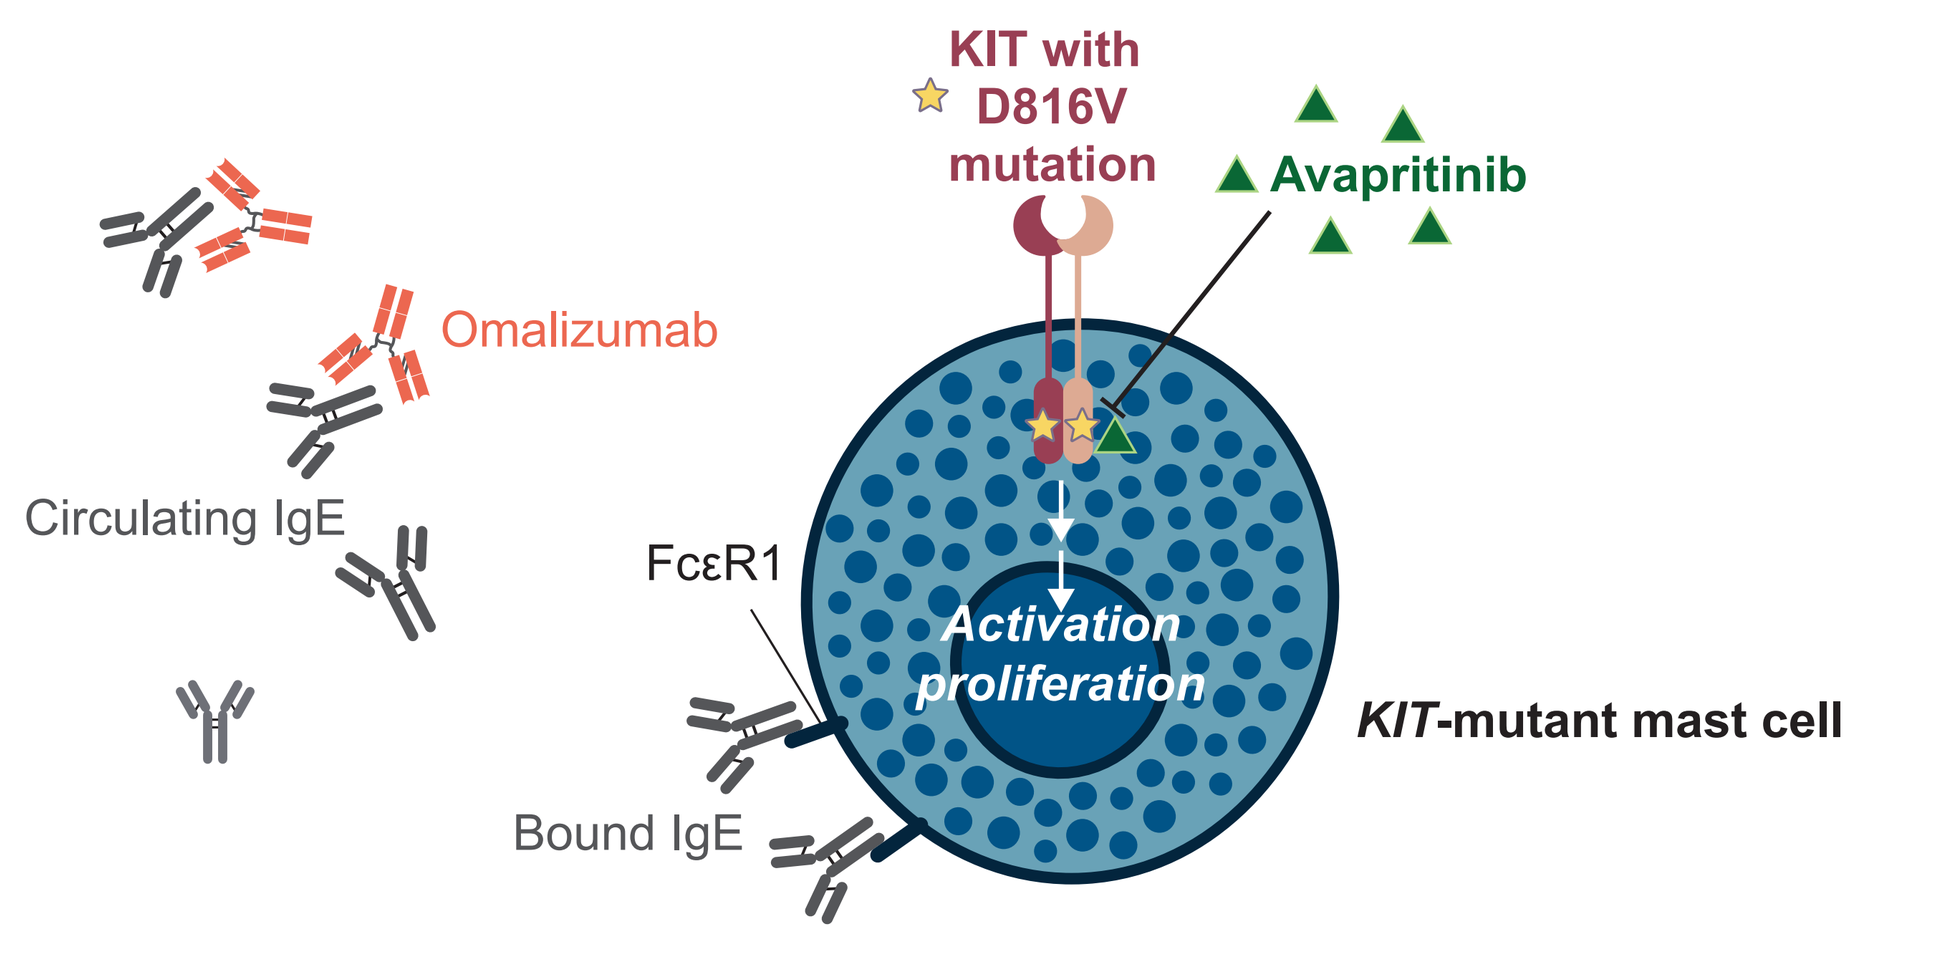
KIT with D816V mutation
Avapritinib
Omalizumab
Circulating IgE
FcεR1
Activation proliferation
KIT-mutant mast cell
Bound IgE

- Patients with ISM who experience anaphylaxis are often treated in real-world clinical settings with the anti-IgE antibody omalizumab as symptom-directed best supportive care (BSC)
 - Omalizumab is not approved for ISM and does not reduce ISM-related biomarkers (e.g., tryptase), although in case series it has demonstrated some benefit in patients with recurrent anaphylaxis[12–14]
- The PIONEER trial (NCT03731260) was a phase 2, randomized, double-blind, placebo-controlled study of avapritinib, a potent, oral inhibitor that selectively targets KIT D816V[15]
 - Avapritinib is now approved in adult patients with ISM in the US and in patients with moderate-to-severe ISM in the EU[16,17]
 - In PIONEER, treatment with omalizumab was allowed as BSC
- Here, we assessed mast cell burden and IgE levels in patients with ISM enrolled in PIONEER, stratified by concomitant omalizumab use and compared to age-matched healthy controls

Methods

- Patients with moderate-to-severe ISM with symptoms that were inadequately controlled despite BSC were enrolled in the randomized, double-blinded, placebo-controlled PIONEER study (**Figure 2**)

Figure 2. Study design

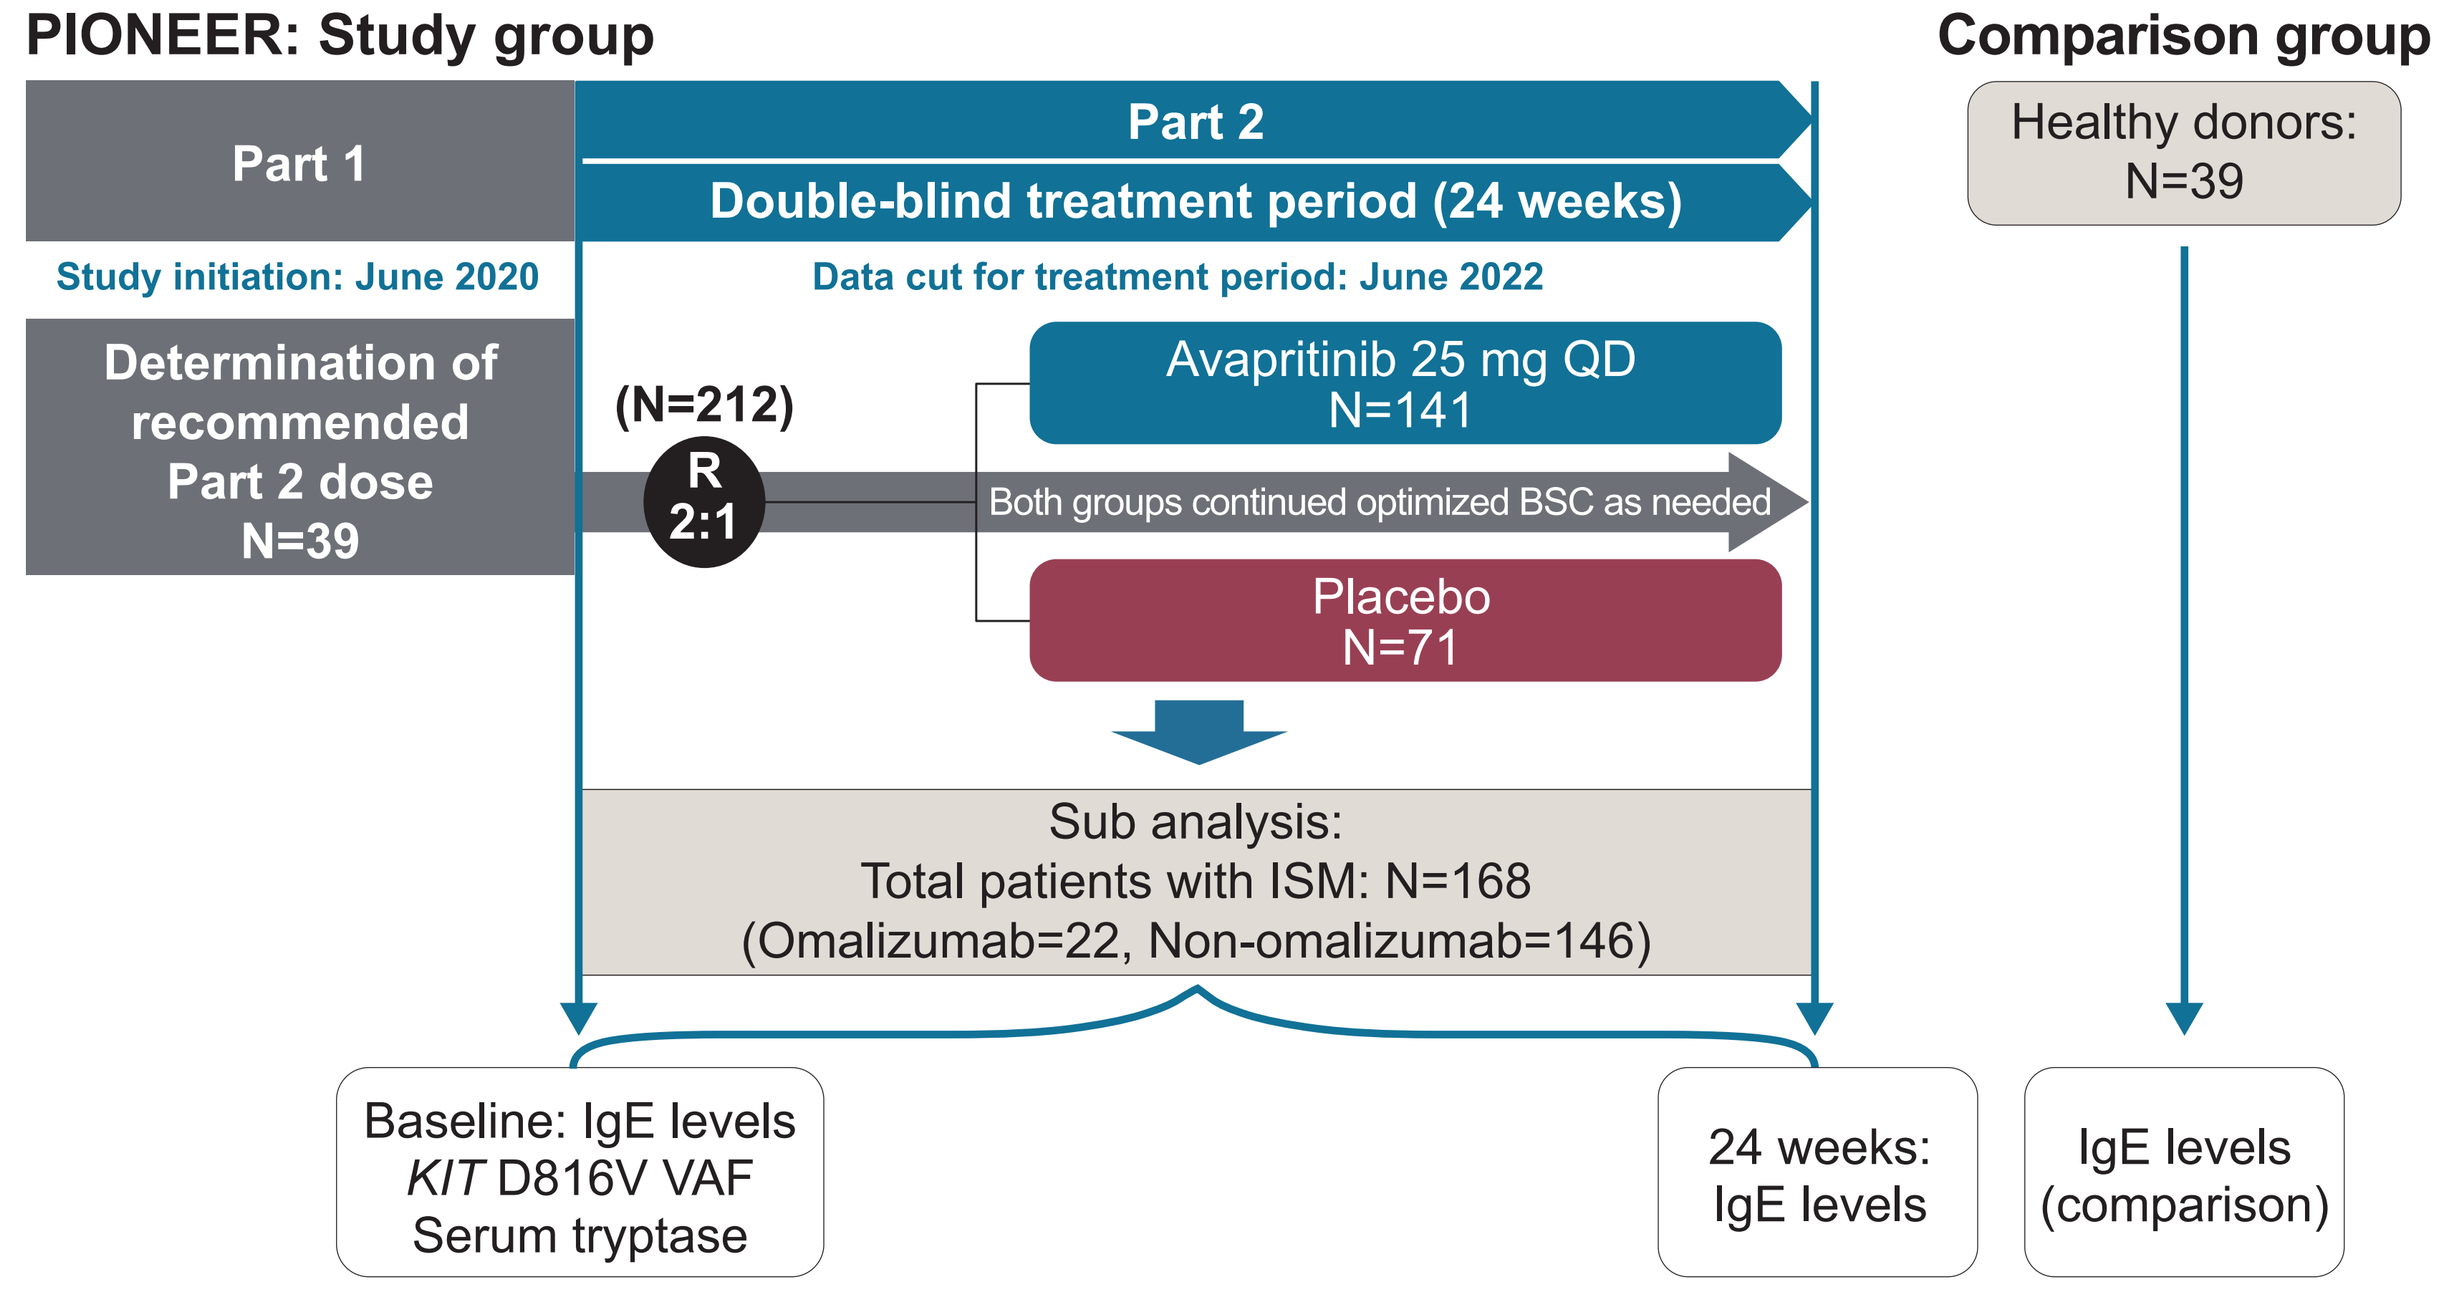
PIONEER: Study group
Comparison group
Part 1
Part 2
Double-blind treatment period (24 weeks)
Study initiation: June 2020
Data cut for treatment period: June 2022
Determination of recommended Part 2 dose N=39
(N=212)
R 2:1
Avapritinib 25 mg QD N=141
Both groups continued optimized BSC as needed
Placebo N=71
Healthy donors: N=39
Sub analysis:
Total patients with ISM: N=168
(Omalizumab=22, Non-omalizumab=146)
Baseline: IgE levels
KIT D816V VAF
Serum tryptase
24 weeks: IgE levels
IgE levels (comparison)

BSC, best supportive care; IgE, immunoglobulin E; QD, once daily; R, randomized; VAF, variant allele frequency.

- In Part 2 of the study, 212 patients were treated with either avapritinib (25 mg orally once daily) or placebo, both with BSC, for 24 weeks[15]
- Plasma samples were collected from patients with ISM and age-matched healthy donors at baseline and after 24 weeks of treatment
 - Total circulating IgE levels in patient plasma were determined at Virant Diagnostics (Wheaton, Maryland, USA) using the FDA-cleared, non-modified method (ImmunoCAP™ Total IgE Fluoroenzyme immunoassay) on Phadia 250 instrument (Thermo Fisher Scientific, Waltham, MA)
 - Total IgE concentrations in samples were automatically calculated from a standard curve using a log-log regression with Phadia Information Data Manager Software
 - Healthy donor plasma samples were collected by Discovery Life Sciences
 - Statistical analyses for IgE level comparison among different groups were done by ordinary one-way ANOVA (Dunnett's multiple comparisons test) or between two groups by unpaired t test (two-tailed) with Welch's correction, and the Pearson correlation coefficient was calculated using GraphPad Prism software
- *KIT* D816V variant allele frequency (VAF; per droplet digital polymerase chain reaction) and serum tryptase were determined at baseline

Results

Patient Demographics

- Samples were collected from 168 patients with ISM from PIONEER at baseline, and from 39 age-matched healthy donors
 - In the group of PIONEER patients with samples available for IgE analysis, 109 (65%) received avapritinib and 59 (35%) received placebo in Part 2

Table 1. Baseline characteristics

Patient characteristic	Patients with ISM who had IgE levels tested (N=168)
Median age, years (range)	51.5 (22–79)
Female, n (%)	126 (75)
ISM-SAF TSS, mean (range)	51.40 (12.1–104.4)
Median serum tryptase, ng/mL (range)	43.60 (3.6–501.6)
Median KIT VAF in PB, % (range)[a]	0.395 (0.00–41.29)
Median BM MCs, % (range)	7.00 (1.0–70.0)
Prior cytoreductive therapy[b], n (%)	20 (12)

[a]By droplet digital polymerase chain reaction; limit of detection 0.02%. [b]Cytoreductive therapies included imatinib, masitinib, dasatinib, midostaurin, nilotinib, cladribine, hydroxyurea, interferon alpha, rapamycin, and brentuximab vedotin.
BM MCs, bone marrow mast cells; ISM, indolent systemic mastocytosis; ISM-SAF, Indolent Systemic Mastocytosis Symptom Assessment Form (© 2018 Blueprint Medicines Corporation); PB, peripheral blood; TSS, total symptom score.

- Of the 168 patients with ISM, 22 (13%) were receiving omalizumab at baseline, and 146 (87%) were not receiving omalizumab at baseline
 - Of the patients who received omalizumab, 17 (77%) received avapritinib and 5 (23%) received placebo in Part 2
- Baseline characteristics are available in **Table 1**

Baseline circulating IgE levels

Figure 3. Circulating total IgE levels at baseline in patients with ISM (A) not receiving omalizumab or (B) receiving omalizumab compared with healthy donors

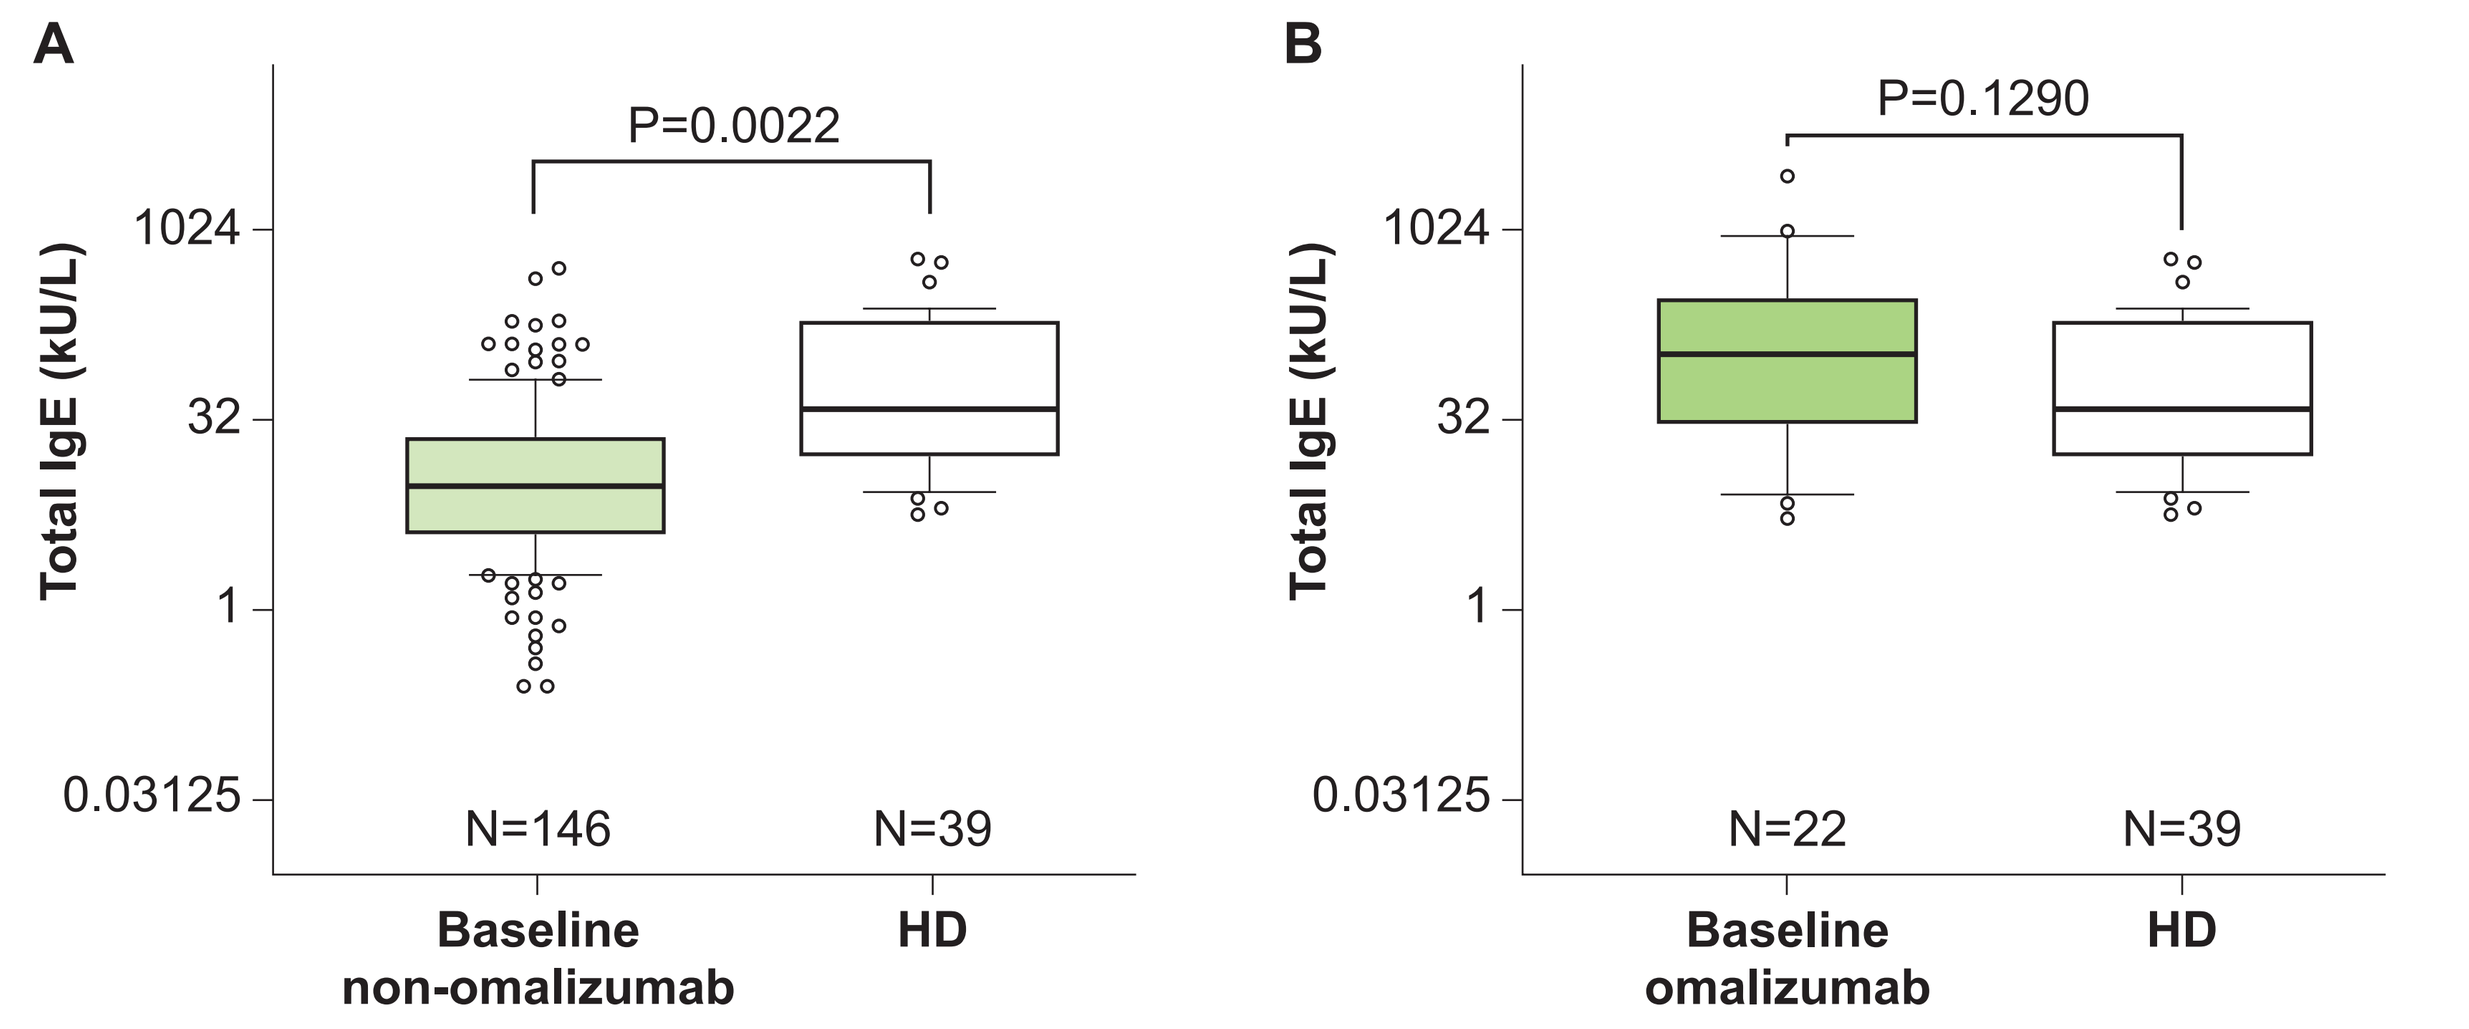
A
P=0.0022
Total IgE (kU/L)
1024
32
1
0.03125
N=146
N=39
Baseline non-omalizumab
HD
B
P=0.1290
Total IgE (kU/L)
N=22
N=39
Baseline omalizumab
HD

	Baseline total IgE (kU/L)	
	Non-omalizumab	HD
Mean	28.92	106.4
Median (range)	9.60 (0.25–506)	39.0 (5.68–596)

	Baseline total IgE (kU/L)	
	Omalizumab	HD
Mean	307.48	106.4
Median (range)	105.30 (5.28–2704)	39.0 (5.68–596)

HD, healthy donor.

- At baseline, IgE levels for patients not on omalizumab (N=146) were significantly lower (9.60 kU/L [0.25–506.00]), than healthy donors (N=39, 39.0 kU/L [5.68–596.00], P=0.0022; **Figure 3A**)
- Median IgE concentration for patients on omalizumab (N=22) was 105.30 kU/L (5.28–2704.00), which was not significantly different than healthy donors (N=39, 39.00 kU/L [5.68–596.00], P=0.1290; **Figure 3B**)

Figure 4. Correlation between total circulating IgE levels and baseline KIT D816V VAF (top) or tryptase (bottom) in PIONEER patients (A, C) not receiving omalizumab or (B, D) receiving omalizumab

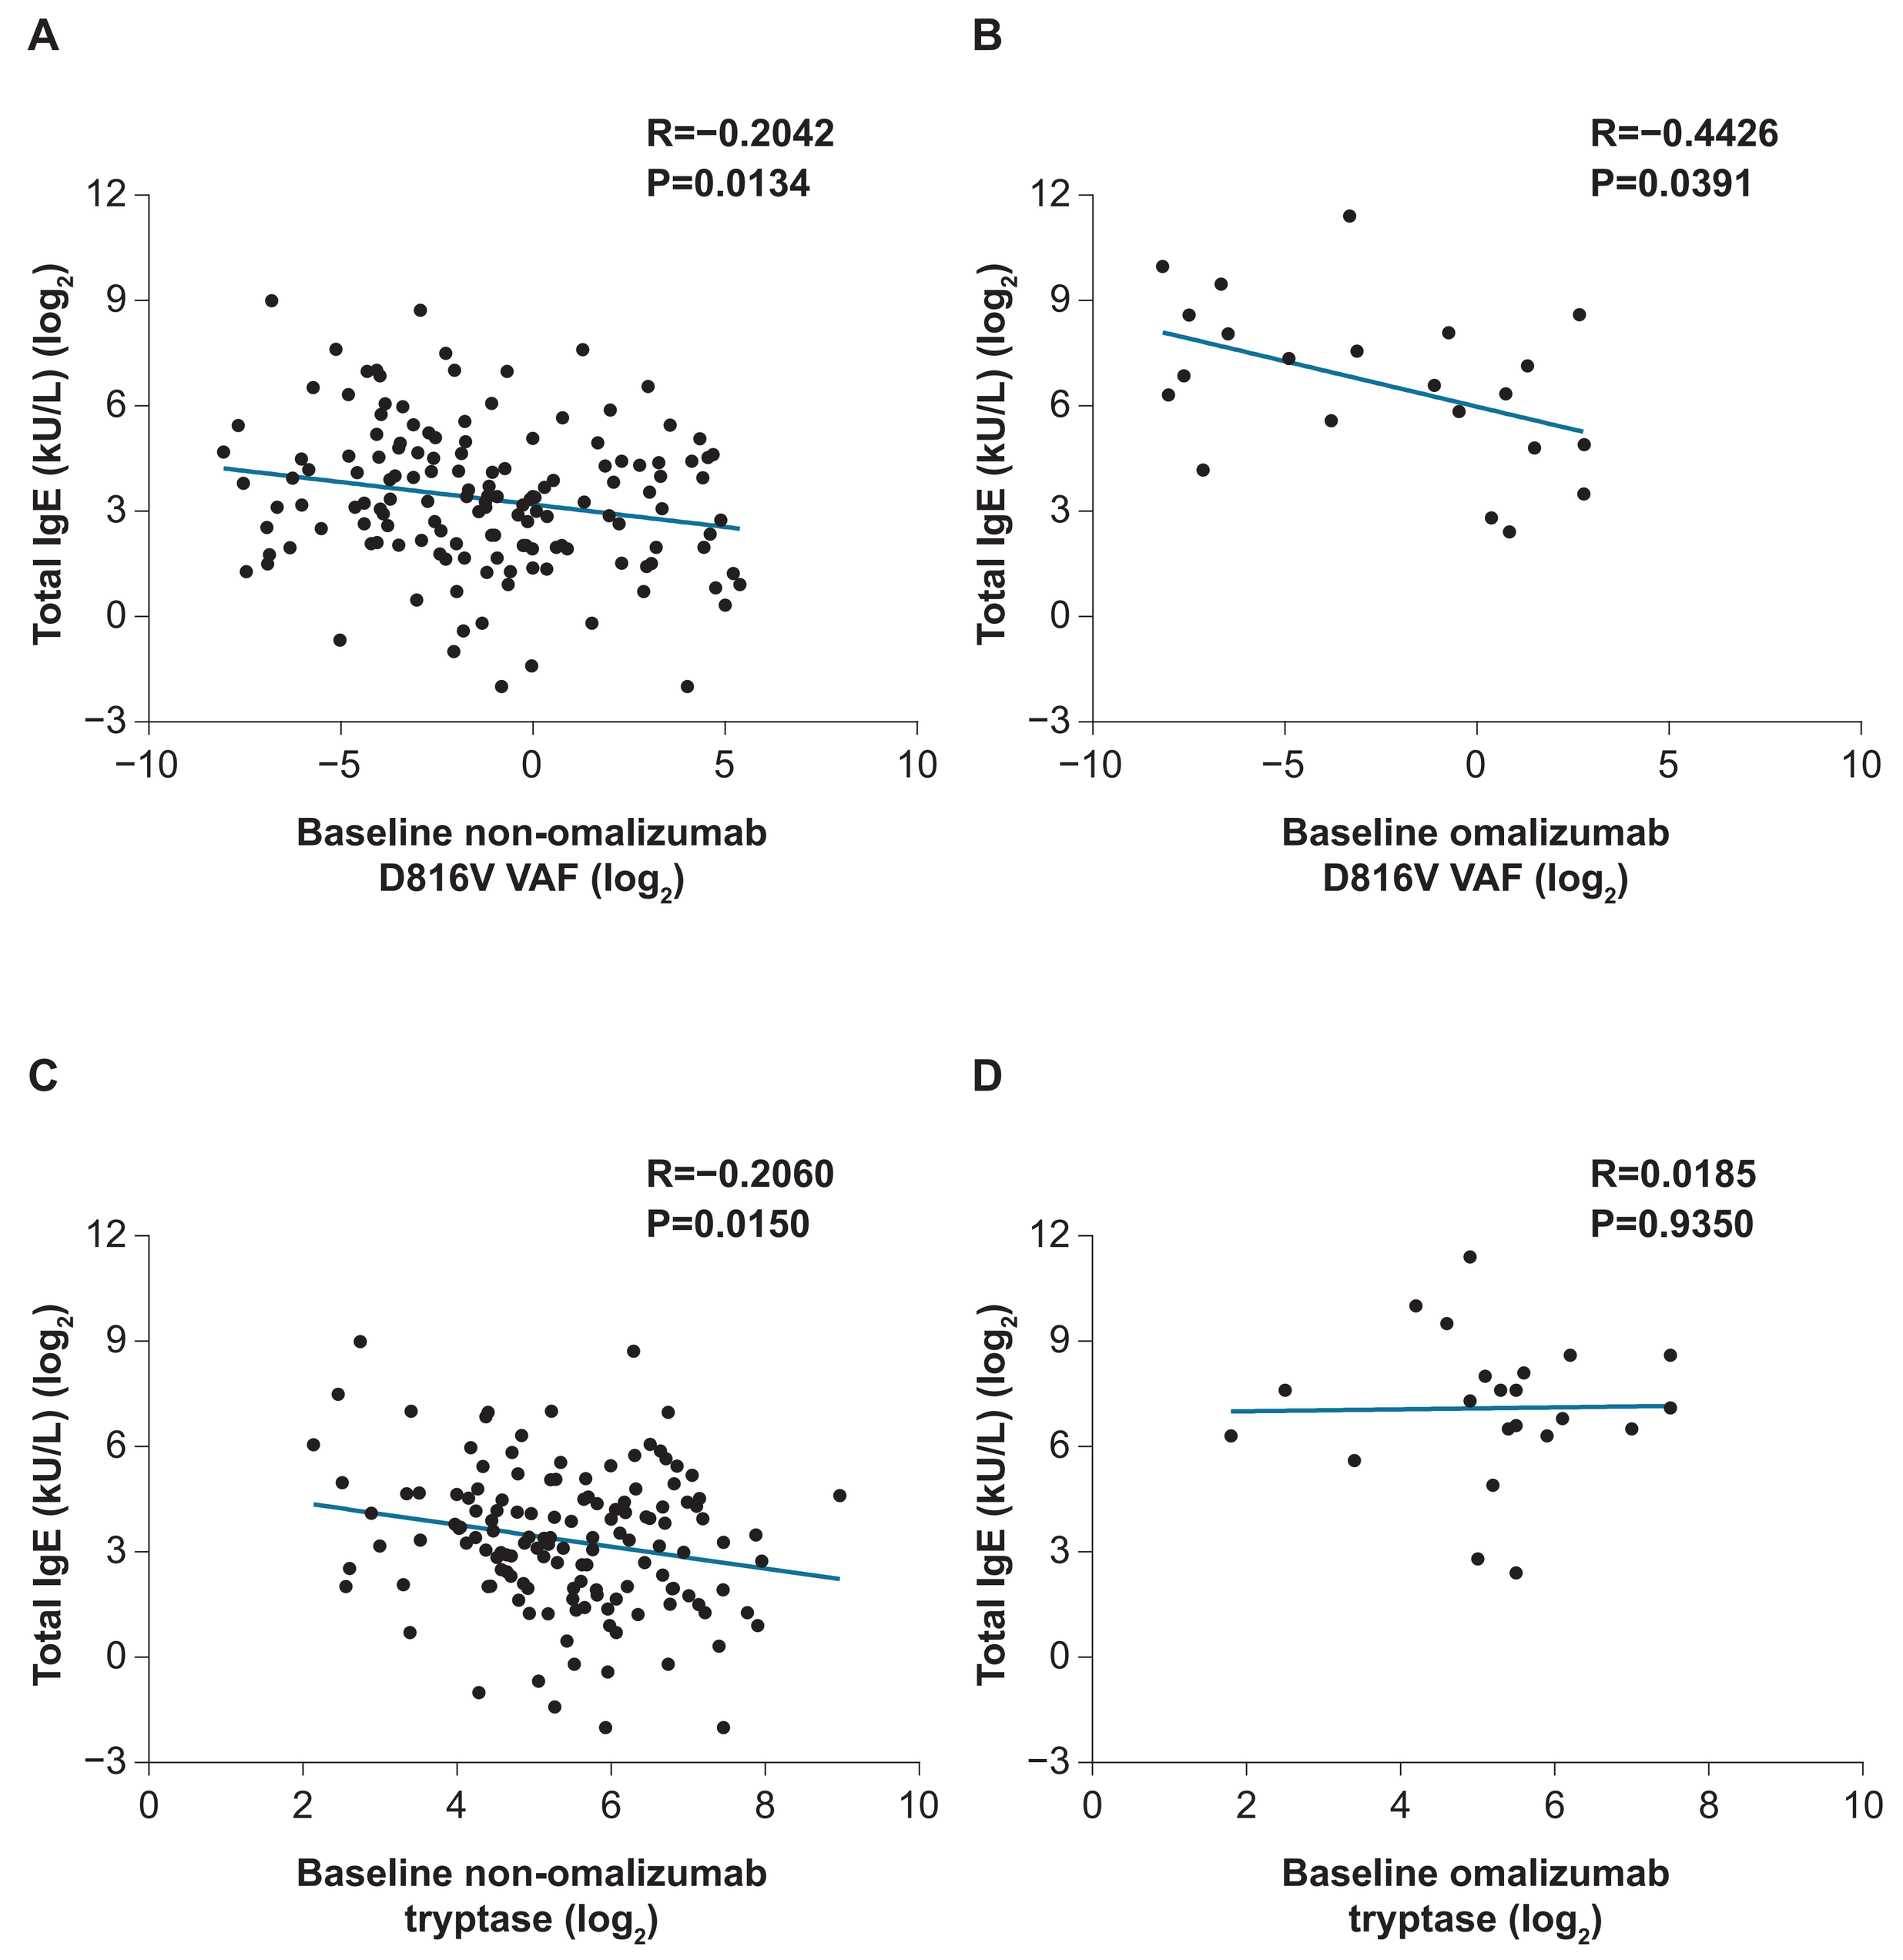
A
R=−0.2042
P=0.0134
Total IgE (kU/L) (log2)
Baseline non-omalizumab D816V VAF (log2)
B
R=−0.4426
P=0.0391
Baseline omalizumab D816V VAF (log2)
C
R=−0.2060
P=0.0150
Baseline non-omalizumab tryptase (log2)
D
R=0.0185
P=0.9350
Baseline omalizumab tryptase (log2)

- There was a negative correlation between IgE levels and *KIT* D816V variant allele frequency in both patients who did not receive omalizumab (R=–0.2042, P=0.0134; **Figure 4A**) and patients who did (R=–0.4426, P=0.0391; **Figure 4B**)
- A negative correlation was observed between baseline IgE and tryptase levels in patients who did not receive omalizumab (R=–0.2060, P=0.0150; **Figure 4C**), but no significant correlation was observed in patients who received omalizumab (R=0.0185, P=0.9350; **Figure 4D**)

References

1. Kristensen T et al. *Am J Hematol*. 2014;89:493–498; 2. Ungerstedt J et al. *Cancers*. 2022;14:3942; 3. Mesa RA et al. *Cancer*. 2022;128:3691–3699; 4. van Anrooij B. et al. *Allergy*. 2016;71:1585–1593; 5. Hartmann K et al. *J Allergy Clin Immunol*. 2016;137:35–45; 6. Brockow K. *Immunol Clin N Am*. 2014;34:283–295; 7. Cohen SS et al. *Br J Haematol*. 2014;166:521–528; 8. van Doormaal JJ et al. *J Allergy Clin Immunol*. 2013;131:1429–1431; 9. Bergström A et al. *Acta Oncol*. 2024;63:44–50; 10. Metcalfe DD. *Blood*. 2008;112:946–956; 11. Gülen T et al. *J Allergy Clin Immunol Pract*. 2017;5:1248–1255; 12. Slapnicar C et al. *Case Rep Hematol*. 2019;6;2019:3787586; 13. Broesby-Olsen S et al. *Allergy*. 2018;73:230–238; 14. XOLAIR (omalizumab) Prescribing Information. San Francisco, CA. Genentech, Inc; 2024; 15. Gotlib J et al. *NEJM Evidence*. 2023;2:EVIDoa2200339; 16. Ayvakit (avapritinib) Prescribing Information. Cambridge, MA: Blueprint Medicines Corporation; 2024; 17. Ayvakyt (avapritinib) Summary of Product Characteristics. Cambridge, MA; Blueprint Medicines Corporation; 2024; 18. Belliveau PP. *MedGenMed*. 2005;7:27; 19. Akin C et al. *Blood*. 2007;1:110:2331–3; 20. Gülen T et al. *Clin Exp Allergy*. 2014;44:1179–1187

Disclosures

Dr. Broesby-Olsen has received honoraria from Blueprint Medicines Corporation, Novartis, and Thermo Fisher; and is a study steering committee member for Blueprint Medicines Corporation.
For all author disclosures, please contact medinfo@blueprintmedicines.com.

Post-treatment circulating IgE levels

Figure 5. Circulating total IgE levels before and after 24 weeks of avapritinib treatment in ISM patients (A) not receiving omalizumab or (B) receiving omalizumab

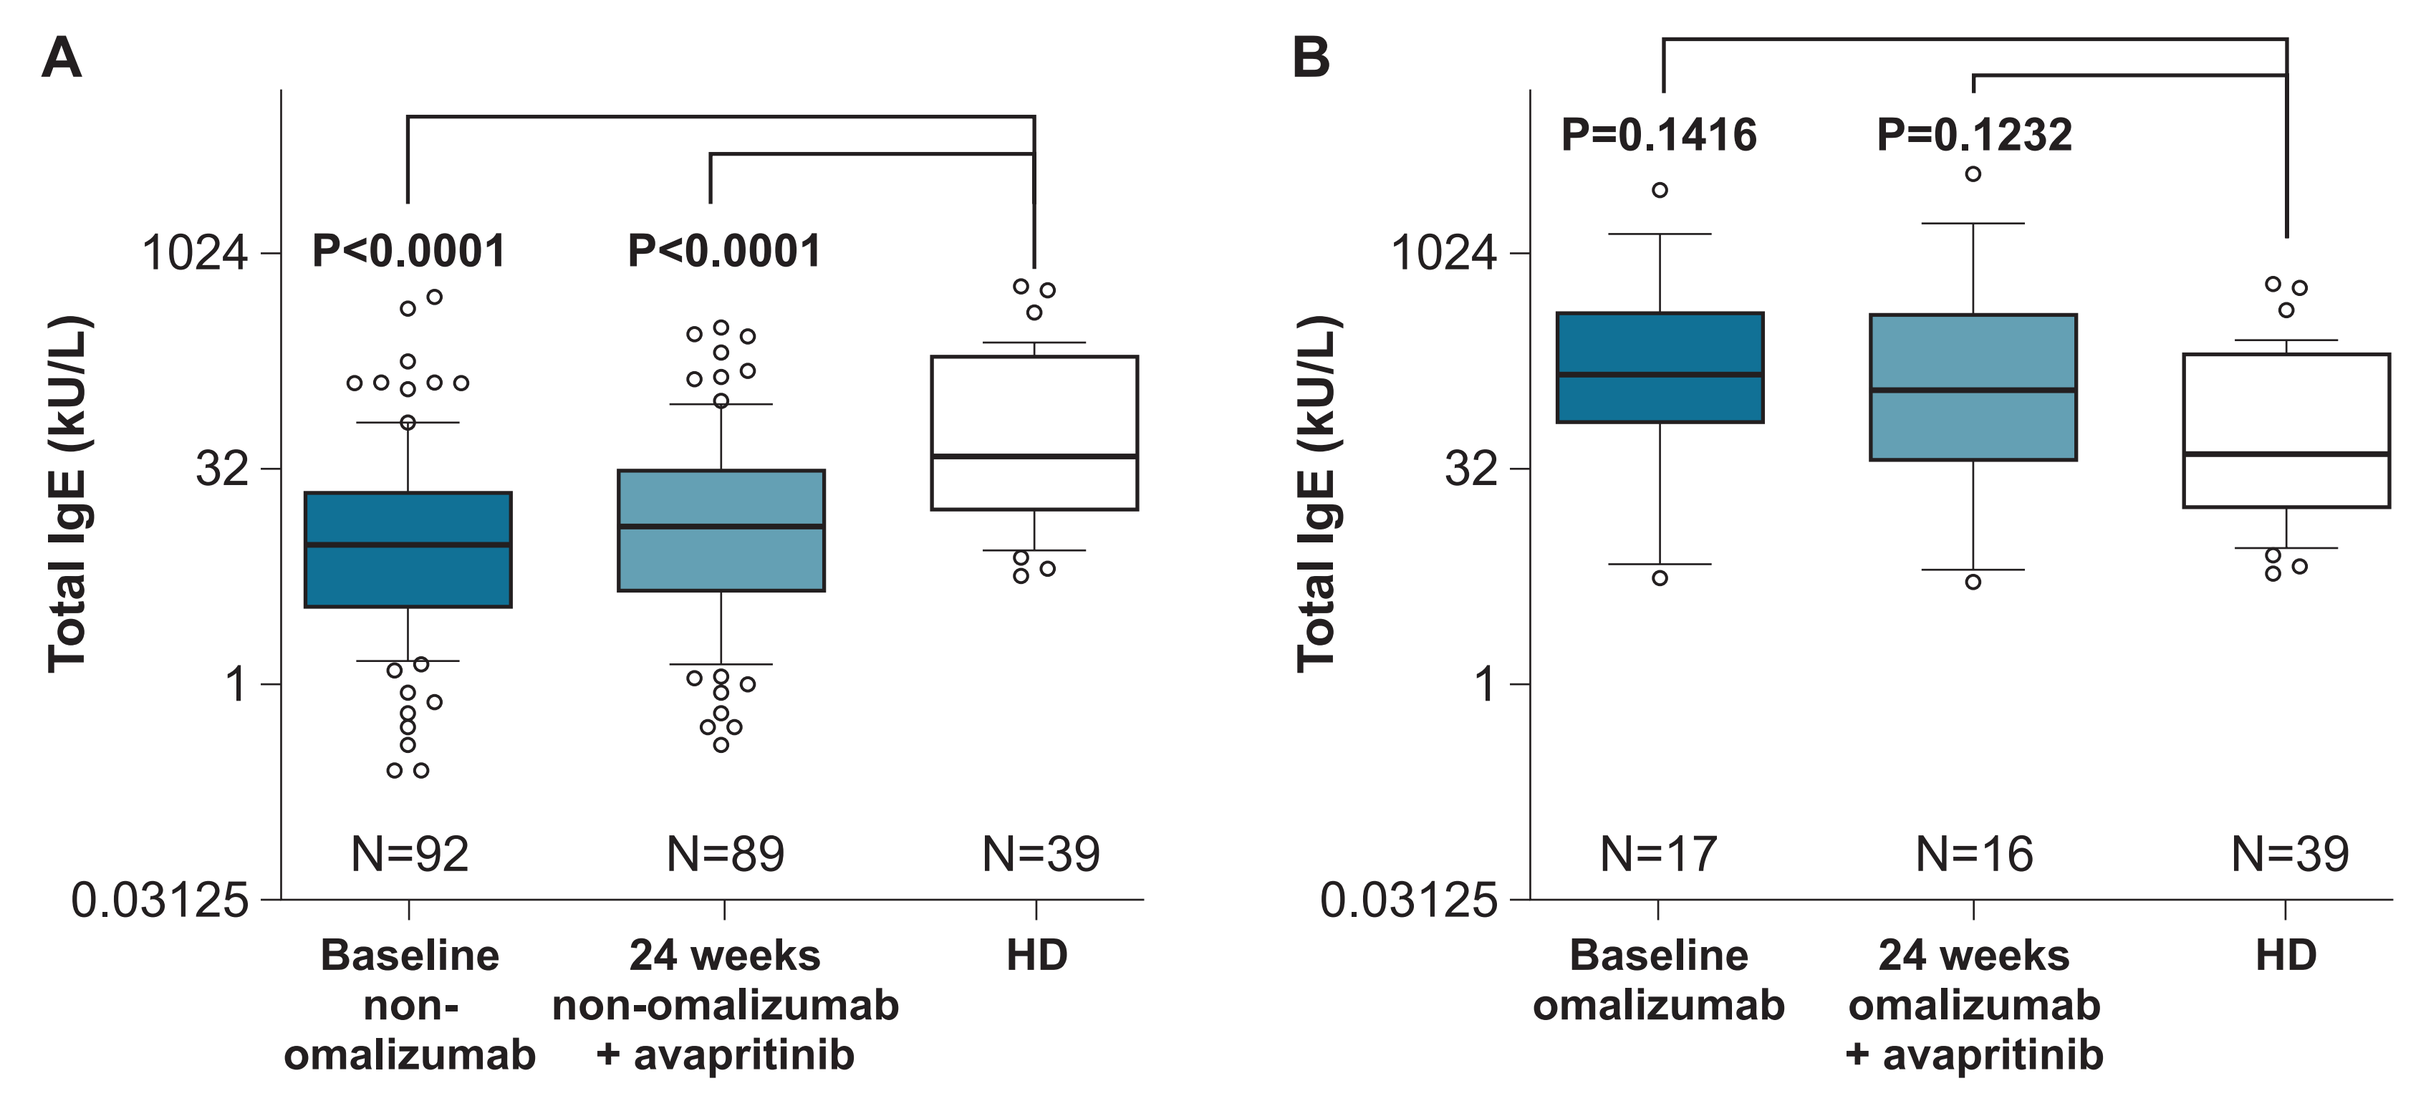
A
P<0.0001
P<0.0001
Total IgE (kU/L)
1024
32
1
0.03125
N=92
N=89
N=39
Baseline non-omalizumab
24 weeks non-omalizumab + avapritinib
HD
B
P=0.1416
P=0.1232
N=17
N=16
N=39
Baseline omalizumab
24 weeks omalizumab + avapritinib
HD

- After 24 weeks of treatment with avapritinib, the levels of IgE trended toward normalized levels in non-omalizumab–treated patients, but were significantly lower than IgE levels in healthy donors (**Figure 5A**)
- The levels of IgE in patients receiving both avapritinib and omalizumab were not significantly changed and were not different than healthy donors (**Figure 5B**)

Conclusions

- This analysis found that patients with ISM who had no prior treatment with omalizumab had significantly lower baseline levels of circulating IgE than healthy donors
 - Due to the formation of omalizumab:IgE complexes upon treatment with omalizumab, elevated serum total IgE levels can be seen for up to a year after treatment with omalizumab, making IgE levels challenging to interpret in this subgroup[14,18]
- We observed a novel inverse correlation between *KIT* D816V VAF and IgE levels in patients with ISM
- We observed an inverse correlation between total IgE and serum tryptase levels in patients not receiving omalizumab, consistent with prior reports[11,19,20]
- An encouraging trend in IgE levels on avapritinib therapy suggests a need for further research to explore whether longer treatment with a KIT D816V inhibitor or the combination of additional immune modulating agents with a KIT D816V inhibitor can achieve normalization of IgE levels in patients with ISM
- Additional research is needed to determine whether the inverse relationship between total IgE levels and mast cell burden is simply due to binding of IgE to the mast cell surface, or whether additional mechanisms are involved
- These findings highlight alterations in the immune effector functions caused by underlying SM

Acknowledgments

We would like to dedicate this work to the memory of our dear colleague, Dr. Marcus Maurer, whose passion, expertise and contribution to the field of allergology and systemic mastocytosis have had a profound impact on patients and colleagues alike. The authors thank the patients, their families, all other investigators and all investigational site members involved in this study. This study was funded by Blueprint Medicines Corporation. Medical writing support was provided by Ashley Knox, PhD, and Travis Taylor, BA, all of Paragon (a division of Prime, Knutsford, UK), funded by Blueprint Medicines Corporation. Responsibility for all opinions, conclusions and data interpretation lies with the authors.

Poster available for download at:

Presented at the 2025 AAAAI/WAO Joint Congress; February 28–March 3, 2025, San Diego, CA. Please contact medinfo@blueprintmedicines.com for permission to reprint and/or distribute.