Abstract 1056

BLU-222, an Investigational, Oral, Potent, and Highly Selective CDK2 Inhibitor, as Monotherapy in Patients With Advanced Solid Tumors and in Combination With Ribociclib and Fulvestrant in HR+/HER2− Breast Cancer

Dejan Juric, MD[1]; Manish R. Patel, MD[2]; Linda R. Duska, MD[3]; Komal Jhaveri, MD, FACP[4]; Brian S. Henick, MD[5]; Ursula A. Matulonis, MD[6]; Pamela N. Munster, MD[7]; Michael J. Birrer, MD, PhD[8]; Kathleen Moore, MD, MS[9]; Giuseppe Curigliano, MD, PhD[10]; Chia-Cheng Li, DDS DMSc[11]; Jian Guo, PhD[11]; Kevin He, PhD[11]; Rentian Wu, PhD[11]; Sam Tamulevich, MS[11]; Mikael L. Rinne, MD, PhD[11]; Timothy A. Yap, MD, PhD[12]

[1]Henri and Belinda Termeer Center for Targeted Therapies, Massachusetts General Hospital, Boston, MA, USA; [2]Florida Cancer Specialists and Sarah Cannon Research Institute, Sarasota, FL, USA; [3]University of Virginia, Charlottesville, VA, USA; [4]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [5]Columbia University Irving Medical Center, New York, NY, USA; [6]Dana-Farber Cancer Institute, Boston, MA, USA; [7]UCSF Helen Diller Family Comprehensive Cancer Center, San Francisco, CA, USA; [8]University of Arkansas for Medical Sciences, Little Rock, AR, USA; [9]Stephenson Cancer Center, Oklahoma City, OK, USA; [10]IEO - Istituto Europeo di Oncologia, Milano, IRCCS, Italy, and Department of Oncology and Hemato-Oncology, University of Milano, Milan, Italy; [11]Blueprint Medicines Corporation, Cambridge, MA, USA; [12]The University of Texas MD Anderson Cancer Center, Houston, TX, USA

Background

- CDK2 is implicated in CDK4/6 inhibitor (CDK4/6i) resistance in hormone receptor–positive/human epidermal growth factor receptor-2–negative (HR+/HER2−) breast cancers.[1] Dependency on CDK2 is also linked to a variety of other aggressive cancers[2]
- CDK2 inhibition combined with CDK4/6i and endocrine therapy is an attractive therapeutic approach to treat or prevent CDK4/6i resistance in HR+/HER2− breast cancers[3,4]
- BLU-222 is an investigational, oral, potent, and highly selective CDK2 inhibitor (CDK2i) with demonstrated activity as monotherapy and combination treatment in preclinical models of HR+/HER2− breast cancers and *CCNE1*–amplified solid cancers[4-6]
- Here, we present clinical data from the ongoing dose–escalation studies of the VELA trial assessing BLU-222 either as monotherapy in patients with heavily pretreated advanced cancers or as combination therapy with ribociclib and fulvestrant in patients with HR+/HER2− breast cancers whose disease has progressed after treatment with a CDK4/6i

Methods

- VELA (NCT05252416) is an international, open–label, first–in–human, phase 1/2 study evaluating the safety, tolerability, pharmacokinetics, pharmacodynamics, and antitumor activity of BLU-222 as monotherapy in adult patients with advanced solid tumors and in combination with ribociclib and fulvestrant for patients with HR+/HER2− breast cancer[7] (**Figure 1**)

Figure 1: Study design

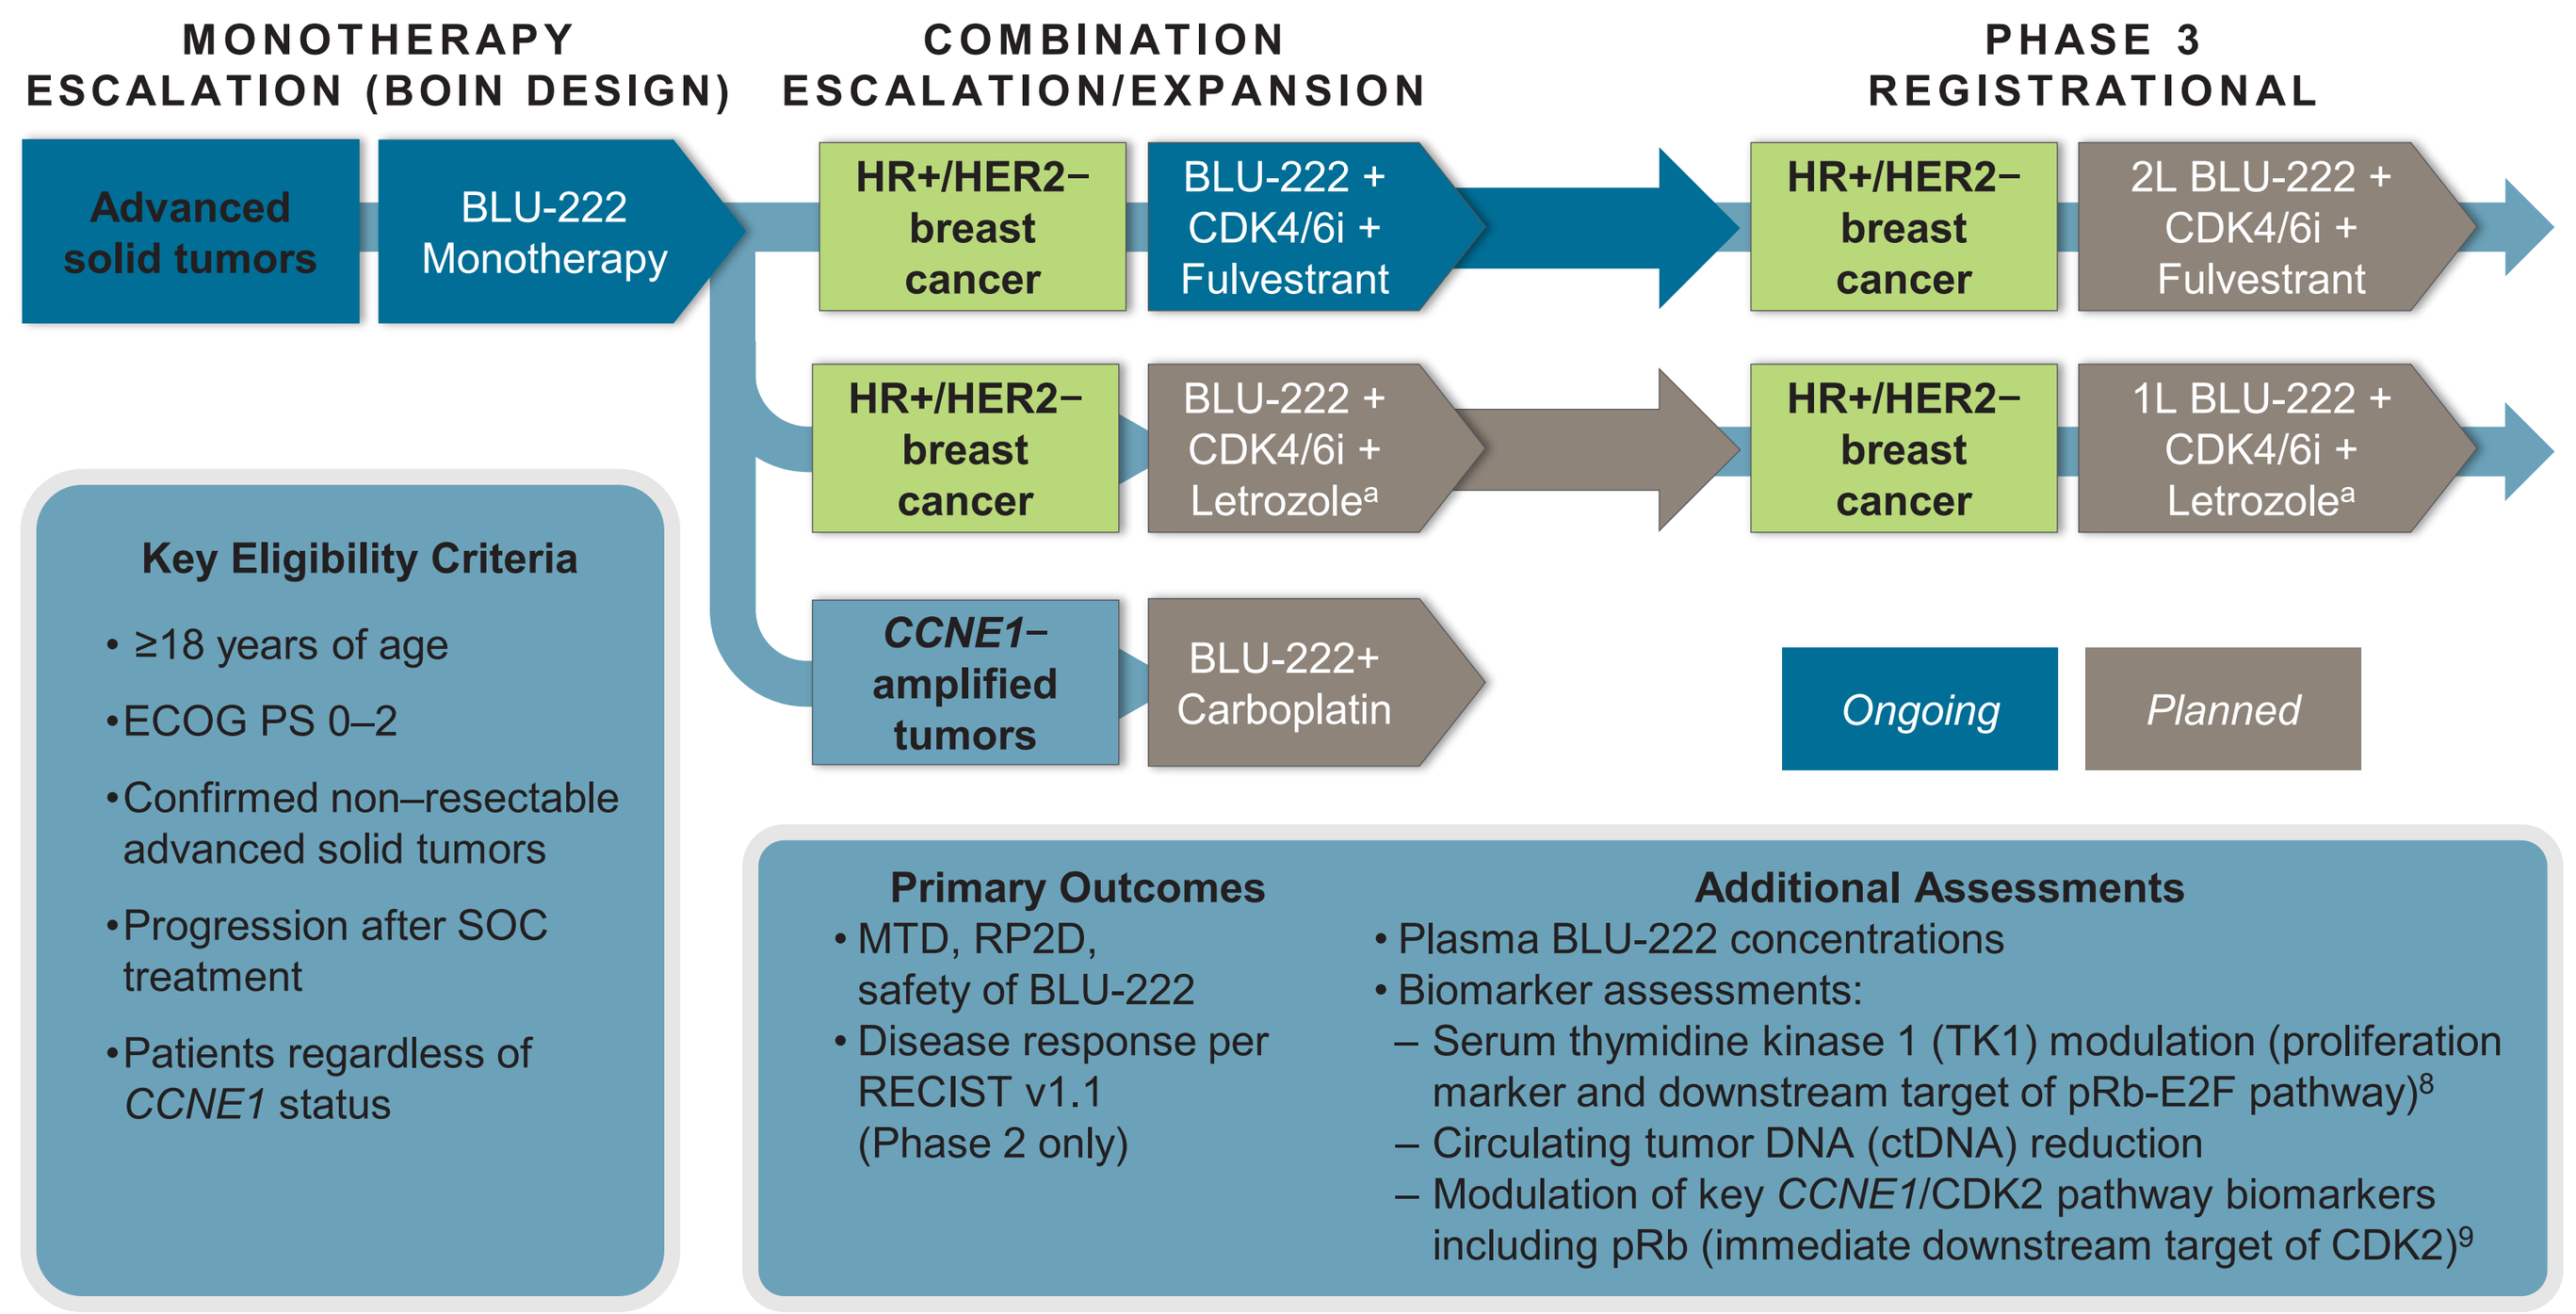

MONOTHERAPY ESCALATION (BOIN DESIGN)
COMBINATION ESCALATION/EXPANSION
PHASE 3 REGISTRATIONAL
Advanced solid tumors
BLU-222 Monotherapy
HR+/HER2− breast cancer
BLU-222 + CDK4/6i + Fulvestrant
HR+/HER2− breast cancer
2L BLU-222 + CDK4/6i + Fulvestrant
HR+/HER2− breast cancer
BLU-222 + CDK4/6i + Letrozole[a]
HR+/HER2− breast cancer
1L BLU-222 + CDK4/6i + Letrozole[a]
CCNE1–amplified tumors
BLU-222+ Carboplatin
Ongoing
Planned
Key Eligibility Criteria
• ≥18 years of age
• ECOG PS 0–2
• Confirmed non–resectable advanced solid tumors
• Progression after SOC treatment
• Patients regardless of CCNE1 status
Primary Outcomes
• MTD, RP2D, safety of BLU-222
• Disease response per RECIST v1.1 (Phase 2 only)
Additional Assessments
• Plasma BLU-222 concentrations
• Biomarker assessments:
– Serum thymidine kinase 1 (TK1) modulation (proliferation marker and downstream target of pRb-E2F pathway)[8]
– Circulating tumor DNA (ctDNA) reduction
– Modulation of key CCNE1/CDK2 pathway biomarkers including pRb (immediate downstream target of CDK2)[9]

[a]Pending protocol amendment, to include first–line metastatic breast cancer patients for dose confirmation.
AE, adverse event; BOIN, Bayesian Optimal Interval; *CCNE1*, cyclin E1; CDK2, cyclin dependent kinase 2; CDK4/6i, cyclin–dependent kinase 4/6 inhibitor; E2F, elongation factor 2; ECOG PS, Eastern Cooperative Oncology Group performance status; HR+, hormone receptor–positive; HER2−, human epidermal growth factor receptor-2–negative; MTD, maximum tolerated dose; pRb, retinoblastoma protein; RECIST, Response Evaluation Criteria in Solid Tumors; RP2D, recommended phase 2 dose; SOC, standard of care.

Results

- As of April 10, 2024, 75 patients were included in the safety population (**Table 1**)
 - 56 in monotherapy cohorts (50−800 mg BLU-222 twice daily [BID])
 - 19 in the combination cohort (100−400 mg BLU-222 BID + 400 mg ribociclib + 500 mg fulvestrant)

Table 1: Demographics and baseline characteristics

	BLU-222 (N=56)	BLU-222 + ribociclib + fulvestrant (N=19)
Age, years, median (min, max)	65 (21, 85)	58 (36, 79)
Age group, years, n (%)		
<65	27 (48.2)	12 (63.2)
≥65	29 (51.8)	7 (36.8)
Sex, n (%)		
Female	47 (83.9)	19 (100)
ECOG PS, n (%)		
0	24 (42.9)	10 (52.6)
1	28 (50.0)	9 (47.4)
2	4 (7.1)	0
Tumor type, n (%)		
Breast	17 (30.4)	19 (100)
Endometrial	5 (8.9)	-
Ovarian	11 (19.6)	-
Uterine	4 (7.1)	-
Other[a]	19 (33.9)	-
Number of prior lines of therapy, median (min, max)	5 (1, 14)	6 (2, 13)[b]
Prior CDK4/6i use (%)[c]	17 (30.4)	19 (100)

[a]Other cancers included prostate (n=3), fallopian tube (n=2), colorectal (n=2), adenocarcinoma, anal neuroendocrine, cervical, chondroblastic osteosarcoma, *CIC-DUX4* sarcoma, extraovarian primary peritoneal clear cell carcinoma, gastric, hepatocellular, pancreatic, lung, thyroid and urothelial cancers (n=1 each).
[b]Prior therapies in metastatic setting, median (min, max): 3 (1, 10).
[c]All HR+/HER2– breast cancer patients were treated with prior CDK4/6i.
CDK4/6i, cyclin–dependent kinase 4/6 inhibitor; ECOG PS, Eastern Cooperative Oncology Group performance status.

BLU-222, ribociclib, and fulvestrant combination therapy

Safety

- BLU-222 (100 mg, 200 mg, and 400 mg) in combination with 400 mg ribociclib and 500 mg fulvestrant has been well tolerated (**Table 2**)
- No patients experienced dose limiting toxicities (DLTs), treatment–related severe adverse events (SAEs), or Grade 4/5 adverse events (AEs)
- Treatment–related hematologic AEs were mostly mild
 - 2 patients experienced Grade 3 white blood cell (WBC) decrease and neutropenia
- Gastrointestinal (GI) AEs were mostly Grade 1 with relatively low rates of diarrhea overall
- 1 patient discontinued treatment (attributed to a ribociclib–related AE[a])

[a]A patient with a history of cardiomyopathy and ventricular tachycardia with an implanted defibrillator who was taken off amiodarone prior to enrollment was taken off study for recurrent ventricular tachycardia.

Table 2: Treatment–emergent adverse events (TEAEs; ≥15%)

	BLU-222 + ribociclib + fulvestrant (N=19)	
Preferred term, n (%)	Any grade	Grade 3[a]
Any TEAEs	19 (100)	7 (36.8)
TEAEs reported in ≥15%		
Nausea	11 (57.9)	0 (0)
Vomiting	6 (31.6)	0 (0)
Anemia	5 (26.3)	2 (10.5)
Blood creatinine increase	5 (26.3)	0 (0)
Diarrhea	4 (21.1)	0 (0)
Neutropenia	4 (21.1)	2 (10.5)
Fatigue	4 (21.1)	0 (0)
Cough	3 (15.8)	0 (0)

[a]No Grade 4 or 5 AEs reported to date.

Pharmacokinetics

- BLU-222 area under the curve (AUC) in combination with ribociclib and fulvestrant increased in a dose–proportional manner across 100, 200, and 400 mg BLU-222 BID combination cohorts (**Figure 2**)
 - Preclinical data demonstrated that efficacy is driven by time over 90% inhibitory concentration (IC_{90}) target coverage (data not shown)
- The average BLU-222 plasma concentration at 400 mg BID in combination cohort covers the projected effective concentration range (C_{eff}) for 10–12 hours of dosing interval
- Ribociclib exposure at steady state was comparable to that reported previously in literature[10]

Figure 2: Plasma concentration–time profiles and AUC after multiple oral administrations of BLU-222 given BID in combination with ribociclib and fulvestrant

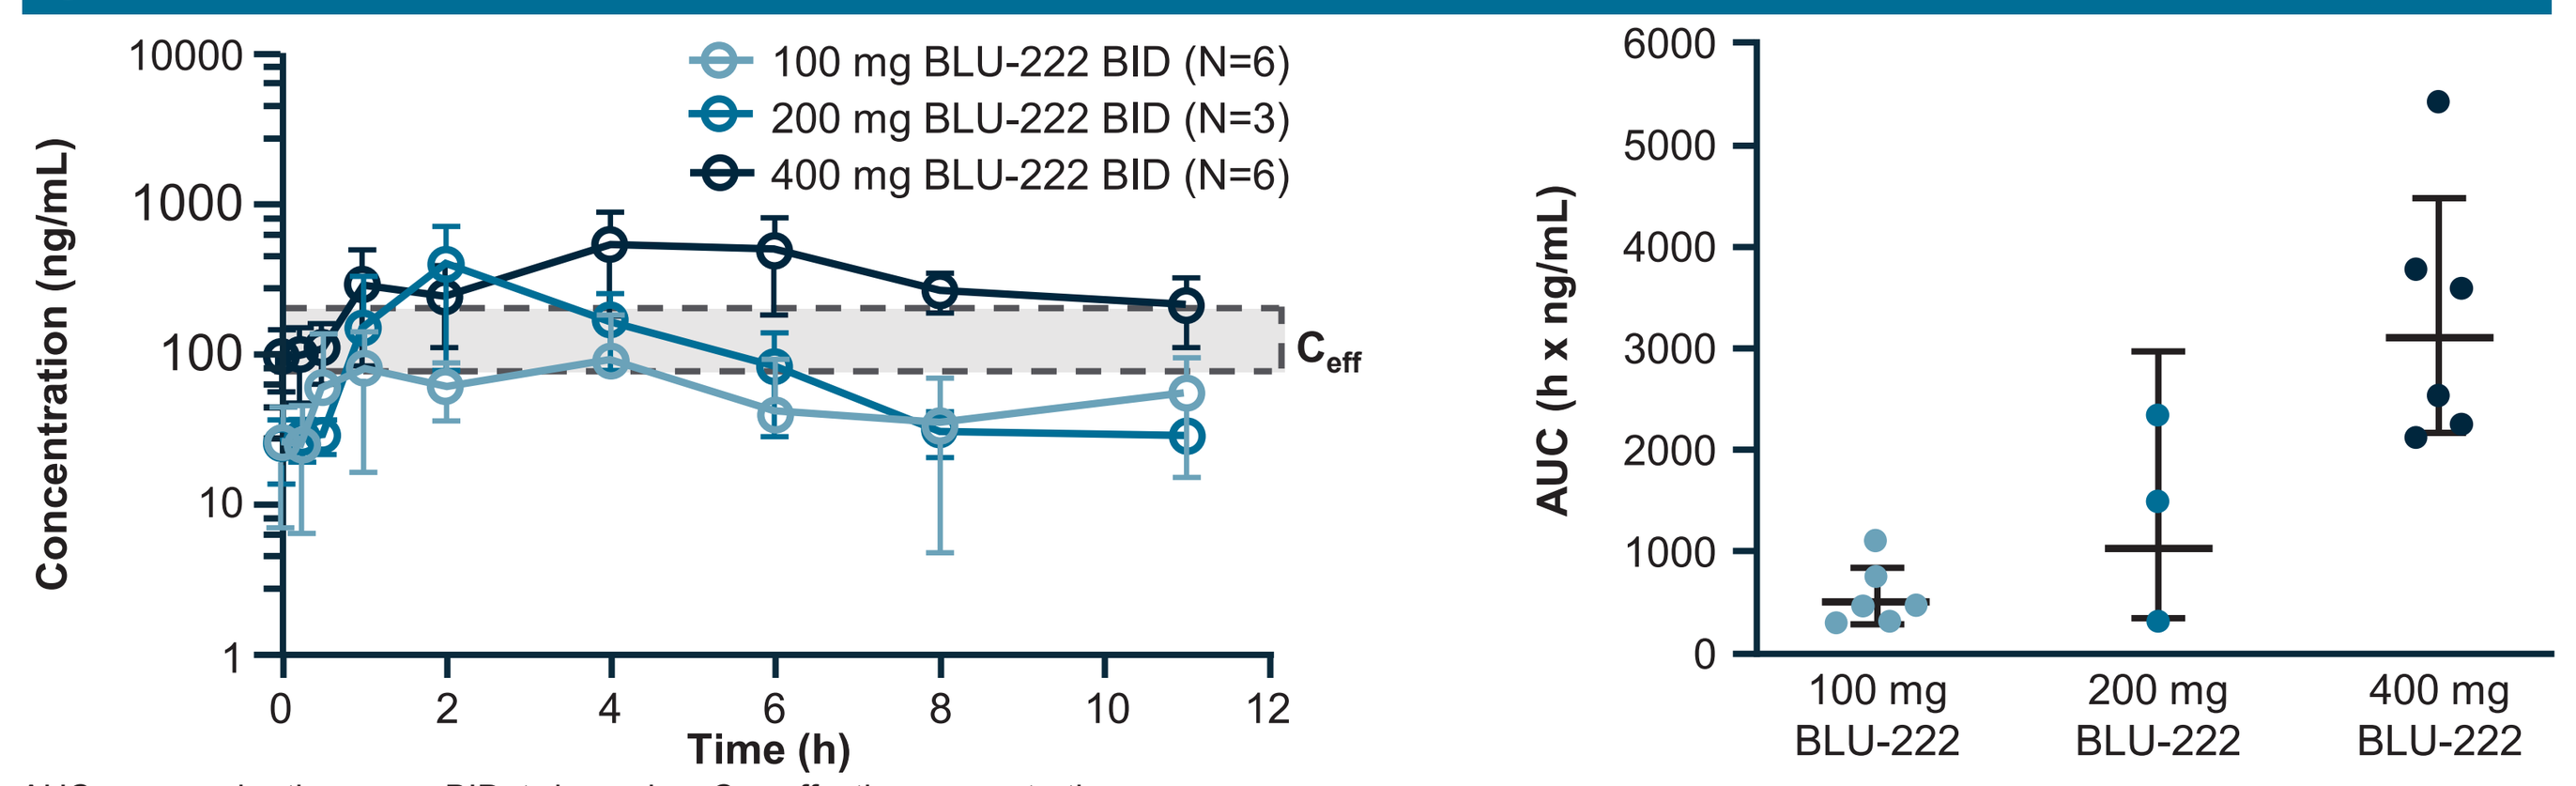

100 mg BLU-222 BID (N=6)
200 mg BLU-222 BID (N=3)
400 mg BLU-222 BID (N=6)
Concentration (ng/mL)
Time (h)
C_{eff}
AUC (h x ng/mL)
100 mg BLU-222
200 mg BLU-222
400 mg BLU-222

AUC, area under the curve; BID, twice a day; C_{eff}, effective concentration.

Pharmacodynamics

- Patients treated at 400 mg BLU-222 + 400 mg ribociclib + 500 mg fulvestrant showed the deepest TK1 reduction (**Figure 3**)
 - TK1 reductions were significantly correlated with increasing exposure to BLU-222 and not ribociclib in patients treated with BLU-222 + ribociclib + fulvestrant combination (**Figure 4**)
- ctDNA reductions were observed in 6/6 patients who had ctDNA data available and were treated with 400 mg BLU-222 + 400 mg ribociclib + 500 mg fulvestrant combination (**Figure 5**)

Figure 3: BLU-222 dose–dependent reductions in TK1 observed in combination with ribociclib + fulvestrant in patients with HR+/HER2− breast cancer

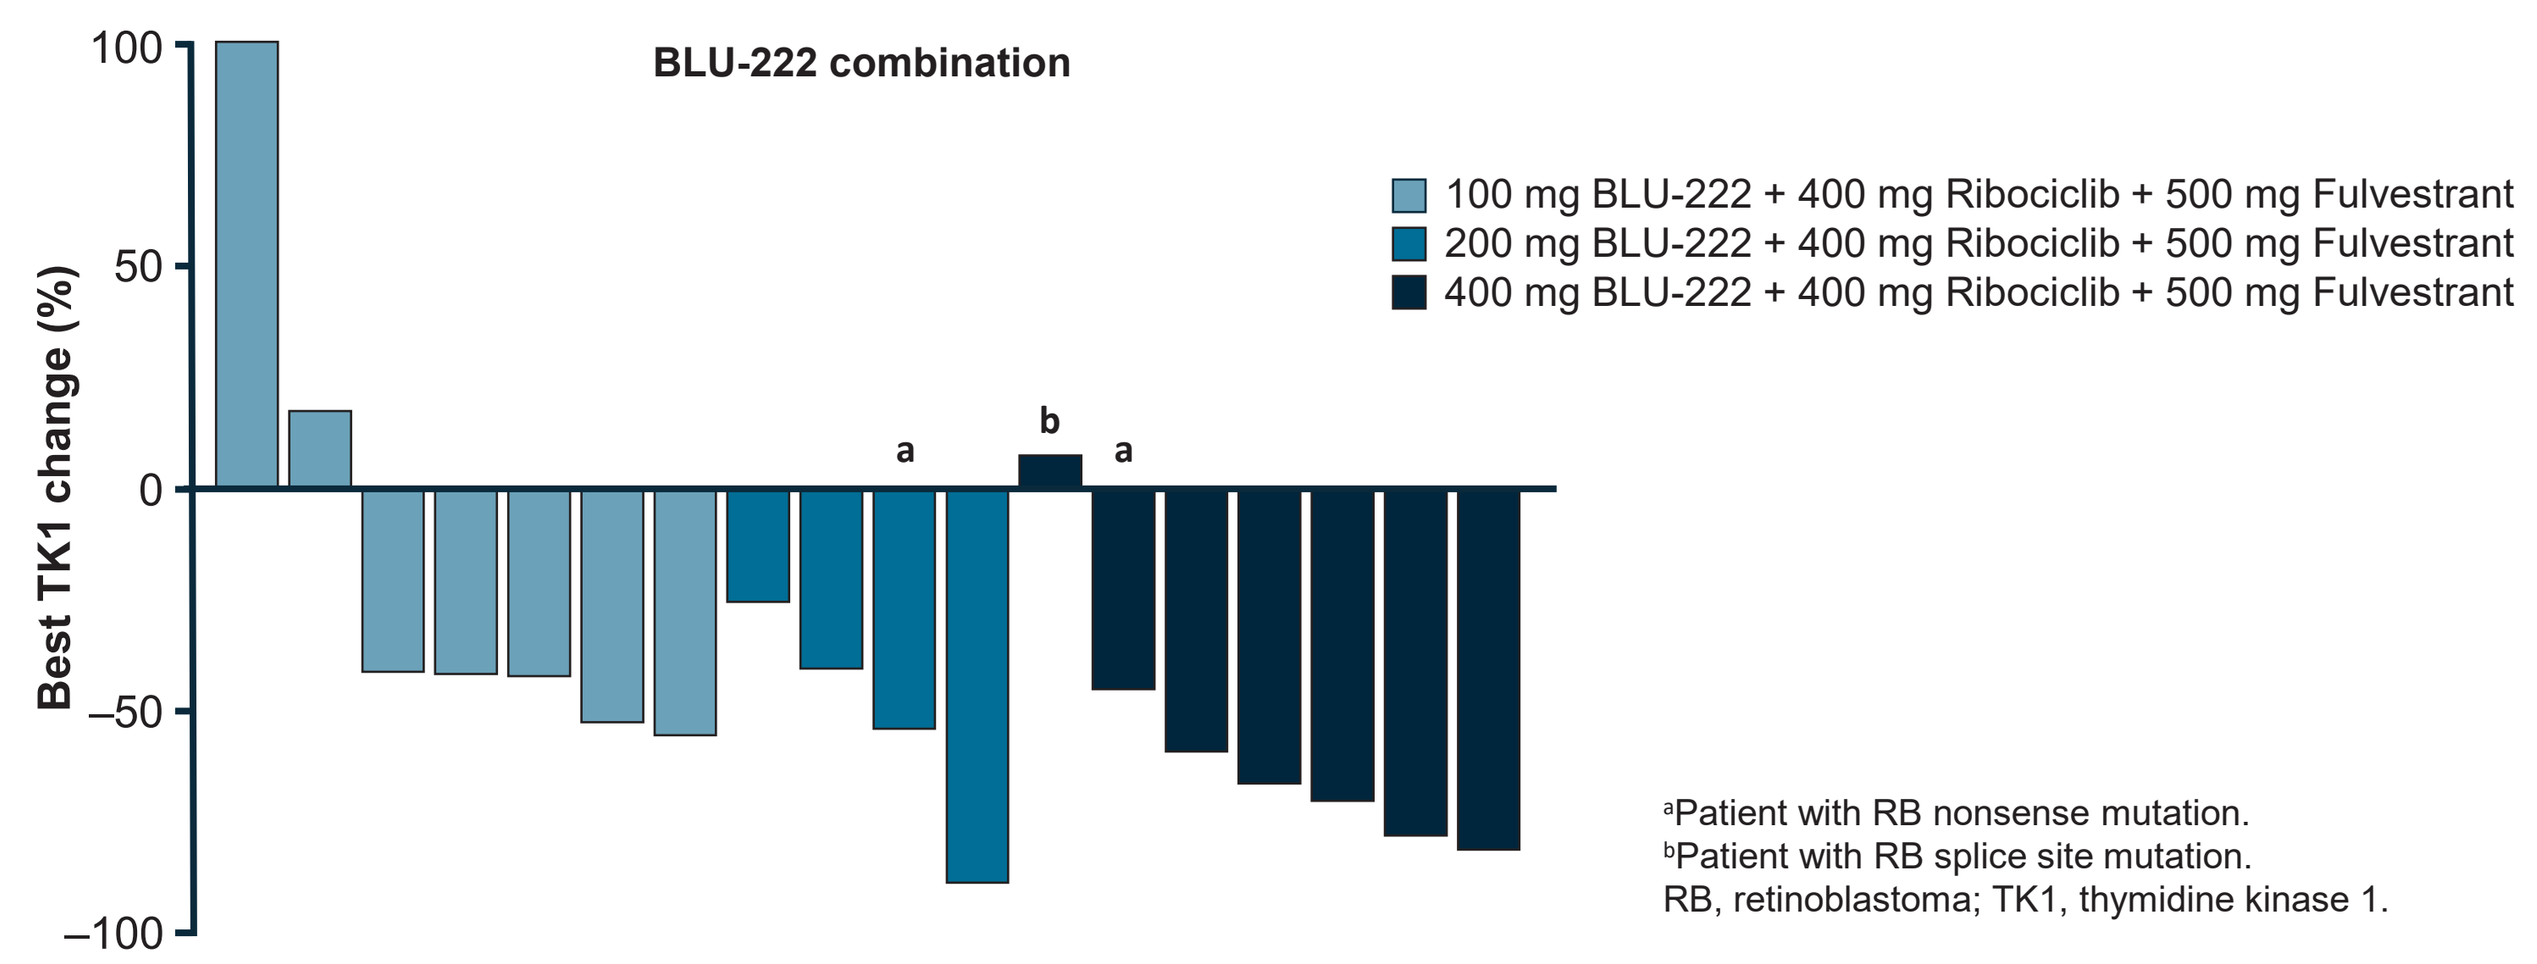

BLU-222 combination
Best TK1 change (%)
100 mg BLU-222 + 400 mg Ribociclib + 500 mg Fulvestrant
200 mg BLU-222 + 400 mg Ribociclib + 500 mg Fulvestrant
400 mg BLU-222 + 400 mg Ribociclib + 500 mg Fulvestrant

[a]Patient with RB nonsense mutation.
[b]Patient with RB splice site mutation.
RB, retinoblastoma; TK1, thymidine kinase 1.

Figure 4: TK1 reductions in patients with HR+/HER2− breast cancer treated with BLU-222 + ribociclib + fulvestrant were significantly correlated with exposure to BLU-222

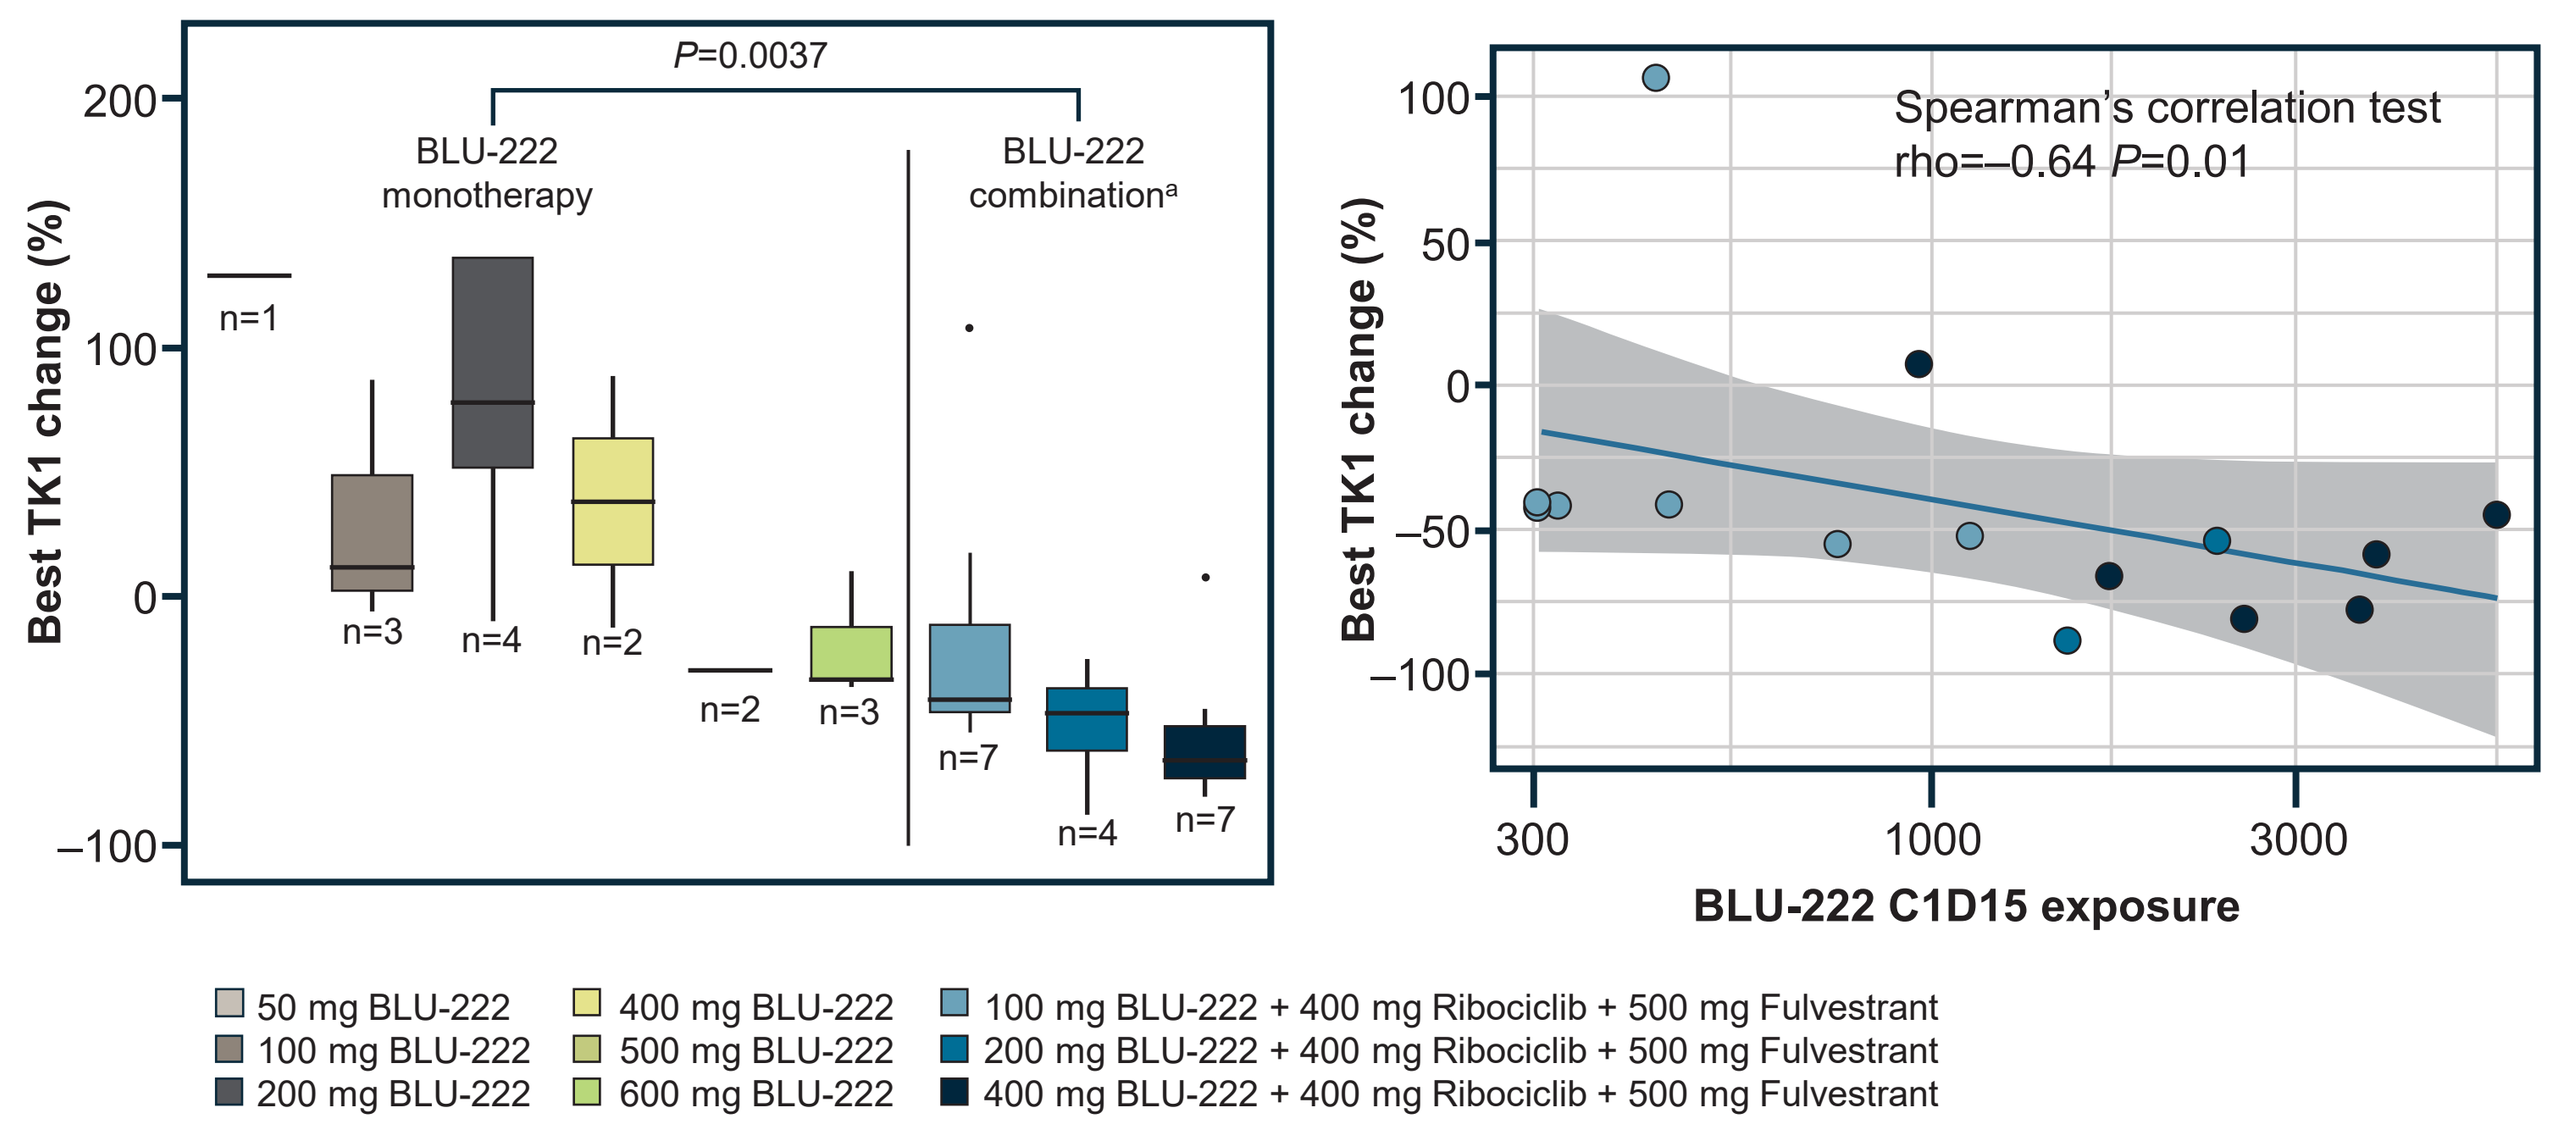

P=0.0037
BLU-222 monotherapy
BLU-222 combination[a]
Best TK1 change (%)
Spearman's correlation test
rho=–0.64 P=0.01
BLU-222 C1D15 exposure
50 mg BLU-222
100 mg BLU-222
200 mg BLU-222
400 mg BLU-222
500 mg BLU-222
600 mg BLU-222
100 mg BLU-222 + 400 mg Ribociclib + 500 mg Fulvestrant
200 mg BLU-222 + 400 mg Ribociclib + 500 mg Fulvestrant
400 mg BLU-222 + 400 mg Ribociclib + 500 mg Fulvestrant

[a]Increased TK1 reduction observed with consistent ribociclib combination dose of 400 mg.
C1D15, cycle 1 day 15; TK1, thymidine kinase 1.

Figure 5: Reduction in ctDNA was observed as measured by both tumor fraction and mVAFR methods in patients treated with BLU-222 + ribociclib + fulvestrant combination

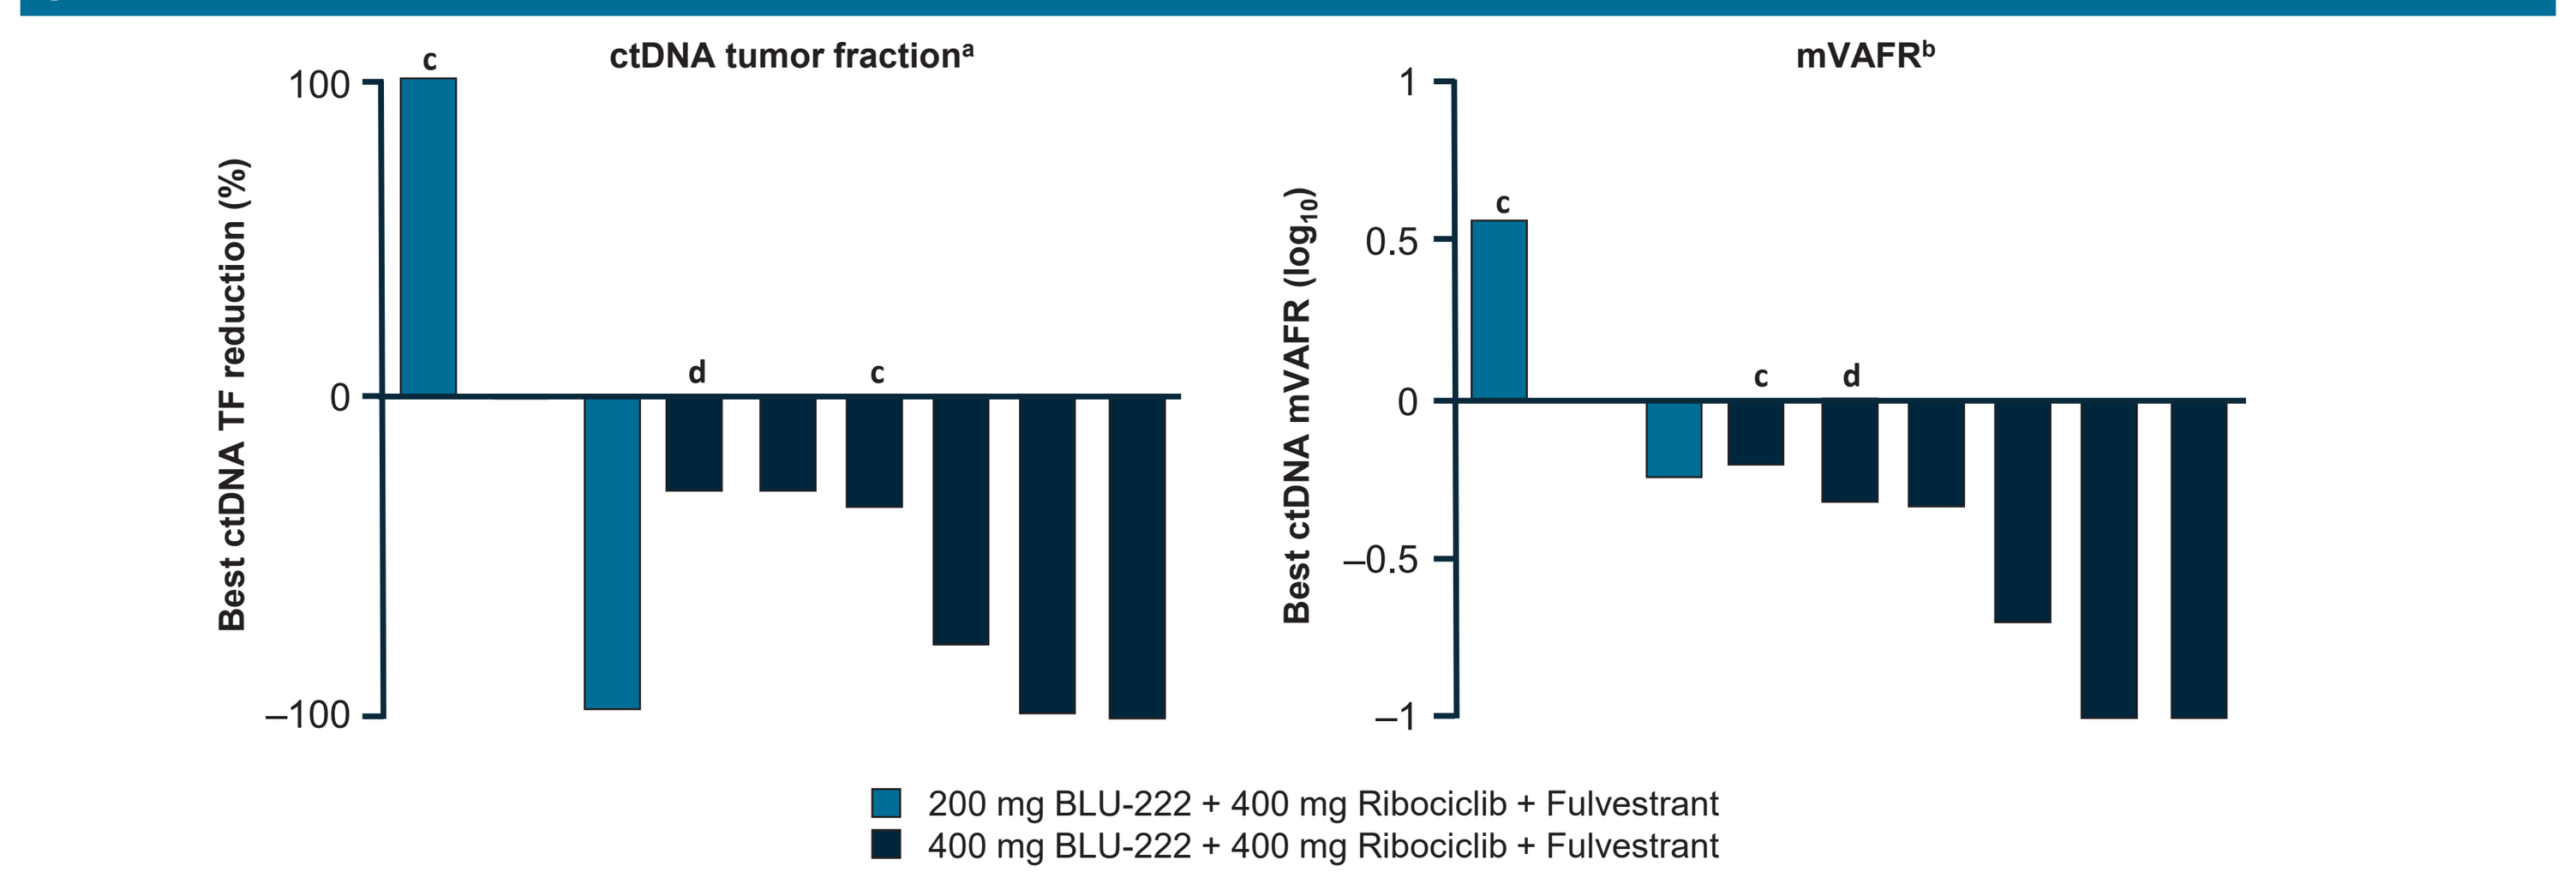

ctDNA tumor fraction[a]
Best ctDNA TF reduction (%)
mVAFR[b]
Best ctDNA mVAFR (log10)
200 mg BLU-222 + 400 mg Ribociclib + Fulvestrant
400 mg BLU-222 + 400 mg Ribociclib + Fulvestrant

[a]Reported as part of FoundationOne®Liquid CDx. ctDNA tumor fraction, quantified by Foundation Medicine's proprietary algorithm, is based on measurements of genomic variation and other Omic information.[11]
[b]Measures proportional change for all cancer–related somatic short variants between baseline (C1D1) and on–treatment timepoints.[12] *DNMT3A*, *TET2*, *JAK2*, *ASXL1*, and *SF3B1* were excluded from analysis due to a high likelihood of clonal hematopoiesis.[13] Variants with VAF ≥40% and < ±10% change between baseline and on–treatment samples were considered germline mutations and excluded from analysis.
[c]Patient with RB nonsense mutation.
[d]Patient with RB splice site mutation.
ASXL1, ASXL transcriptional regulator 1; C1D1, cycle 1 day 1; ctDNA, circulating tumor DNA; *DNMT3A,* DNA methyltransferase 3 alpha; *JAK2*, Janus kinase 2; mVAFR, mean of all variant allele fraction ratios (VAFRs); RB, retinoblastoma; *SF3B1*, splicing factor 3b subunit 1; *TET2*, tet methylcytosine dioxygenase 2; TF, tumor fraction; VAF, variant allele frequency.

Combination therapy patient vignettes

Combination escalation is ongoing. Efficacy in patients treated with BLU-222 in combination with ribociclib and fulvestrant is still immature. Emerging evidence of clinical benefit includes cases of resolution of disease-related bone pain and unconfirmed responses per RECIST criteria in 2 patients after the data cutoff:

Patient vignette 1: partial response (PR, unconfirmed)

77–year–old female with Stage IV HR+/HER2− breast cancer (metastases to bone and liver) with *PIK3CA* and *ESR1* mutations, who had been treated with 6 prior lines of therapy in the metastatic setting (CDK4/6i palbociclib + letrozole, exemestane + everolimus, alpelisib + fulvestrant, capecitabine, gemcitabine, and TDxd). Prior to coming on study, the patient had progression on TDxd with a new liver metastasis

Course of treatment with BLU-222

- Initiated BLU-222 at 400 mg BID in combination with 400 mg ribociclib and 500 mg fulvestrant with no Grade ≥3 AEs reported
- Biomarker results consistent with pathway inhibition and anti-tumor activity:
 - ctDNA tumor fraction was reduced by 29% by C2D1, including reductions in all cancer–associated alleles (**Figure 6**)
 - On-treatment biopsy on C1D10 showed 82% reduction of pRb and 90% reduction of Ki67
 - TK1 activity was reduced by 81% on C1D15
- An unconfirmed PR was observed with a 30% decrease in the target liver lesion after 2 cycles (week 8) of treatment. Treatment is ongoing

Figure 6: ctDNA reduction in patient 1

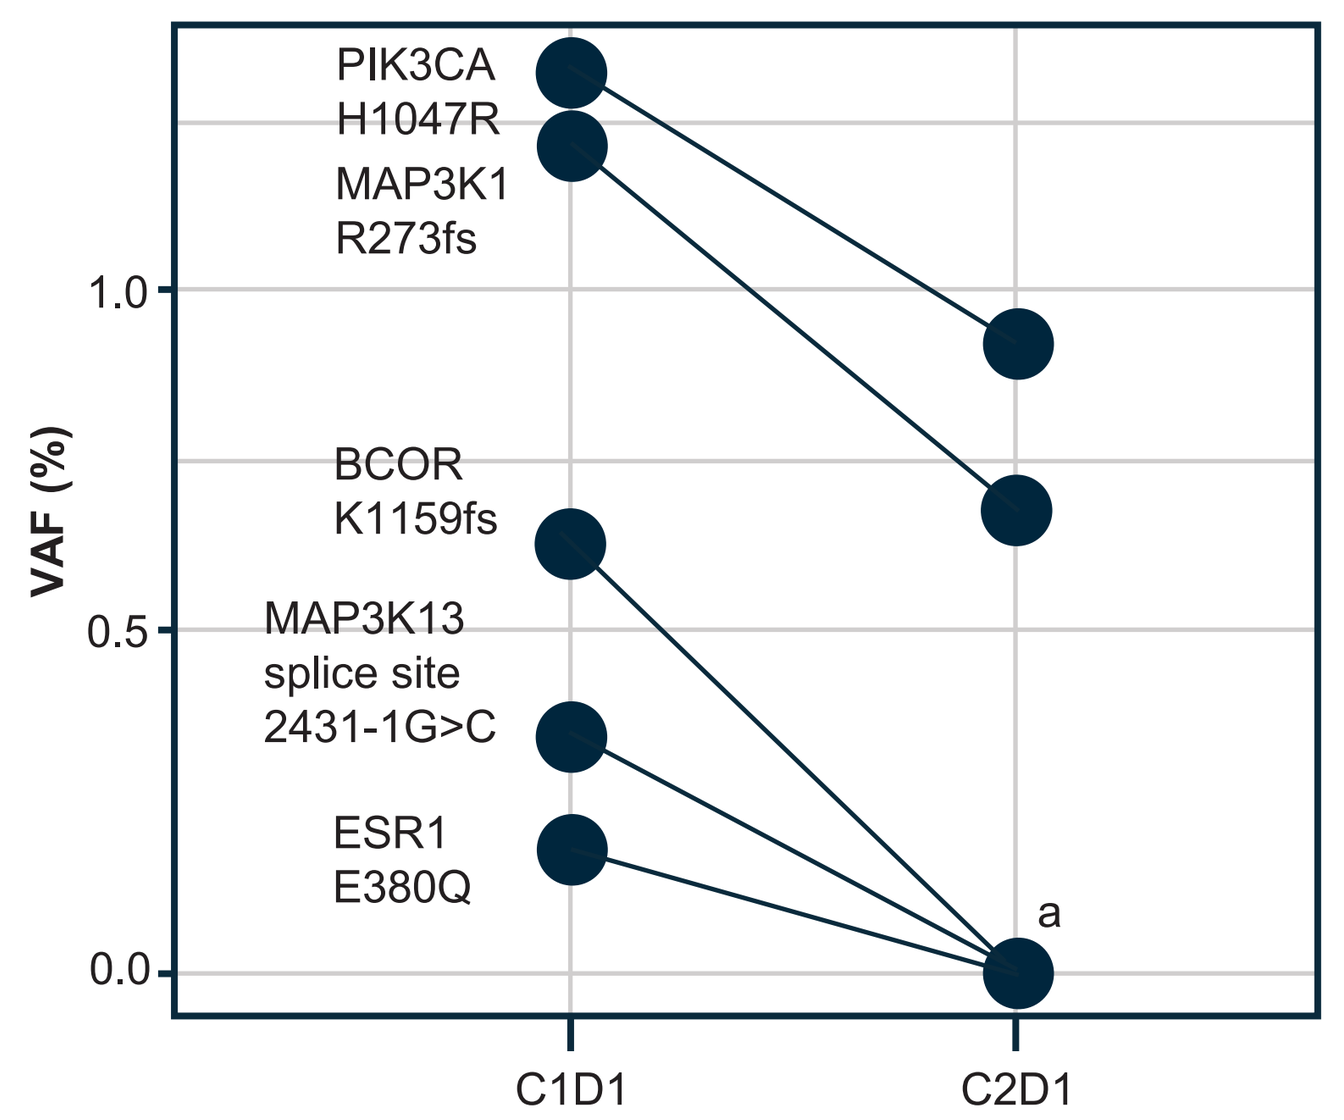

PIK3CA H1047R
MAP3K1 R273fs
BCOR K1159fs
MAP3K13 splice site 2431-1G>C
ESR1 E380Q
VAF (%)
C1D1
C2D1
a

[a]*BCOR*, *MAP3K13* and *ESR1* genomic variations undetectable at C2D1.
BCOR, BCL6 corepressor; ctDNA, circulating tumor DNA; *ESR1*, estrogen receptor 1; *MAP3K*, mitogen–activated protein kinase kinase kinase; *PIK3CA*, phosphatidylinositol-4,5-bisphosphate 3-kinase catalytic subunit alpha; TDxd, trastuzumab deruxtecan; TK1, thymidine kinase 1; VAF, variant allele frequency.

Patient vignette 2: partial response (PR, unconfirmed)

73–year–old female with Stage IV HR+/HER2− breast cancer (metastases to bone, liver, and lung) with *PIK3CA* and *ESR1* mutations, who had been treated with 2 prior lines of therapy in the metastatic setting (CDK4/6i palbociclib + letrozole, mutant–selective PI3K inhibitor RLY2608 + fulvestrant, with progression after just 2 months on study with development of new liver lesions)

Course of treatment with BLU-222

- Initiated BLU-222 at 100 mg BID in combination with 400 mg ribociclib and 500 mg fulvestrant
- Grade 3 neutropenia in Cycle 2 led to ribociclib dose reduction to 200 mg starting on C3D1
- BLU-222 dose was increased to 200 mg BID on C8D1; ribociclib dose remained at 200 mg
- At the beginning of Cycle 9, an unconfirmed PR was observed with the target and non–target lesions in the liver not detected (residual bone disease unchanged). Treatment is ongoing. Patient has been escalated to 400 mg BID BLU-222 starting in Cycle 9

BLU-222 monotherapy

Safety

- BLU-222 monotherapy has been well tolerated to date with no maximum tolerated dose (MTD) identified. Most common treatment–emergent adverse events (TEAEs; ≥15%) were diarrhea (55%), nausea (54%), fatigue (39%), vomiting (38%), anemia (29%), photophobia (18%), hypokalemia (18%). No cardiac AEs were observed in the monotherapy cohort (n=56) and no effect of BLU-222 on QTc was observed in Holter monitoring of a subset of patients (n=12) (**Figure 7**)
- 2 patients experienced DLTs; both improved after dose reduction:
 - Grade 3 nausea in 1 patient at 800 mg BID
 - Grade 3 blurred vision/photophobia in 1 patient at 600 mg BID
- Treatment–related hematologic and GI AEs were mild
- Treatment–related visual AEs[a] were reported in 16 (28.6%) patients
 - Majority were brief, transient/intermittent Grade 1; 1 patient had Grade 3 blurred vision and photophobia. No acute ophthalmologic findings in any patients
- 3 patients discontinued study treatment due to AEs[b]

[a]Biochemical experiments demonstrate that BLU-222 is a weak inhibitor of phosphodiesterase 6 (890 nM PDE6C half–maximal inhibitory concentration [IC_{50}]). Drugs that inhibit PDE6 (i.e., sildenafil and tadalafil) are known to cause transient, reversible visual symptoms such as changes in color or light perception.[14, 15]
[b]2 patients discontinued due to treatment–related AEs (vomiting, nausea).

Figure 7: Holter data in a subset of patients (n=12) suggests no effect of BLU-222 on QTc

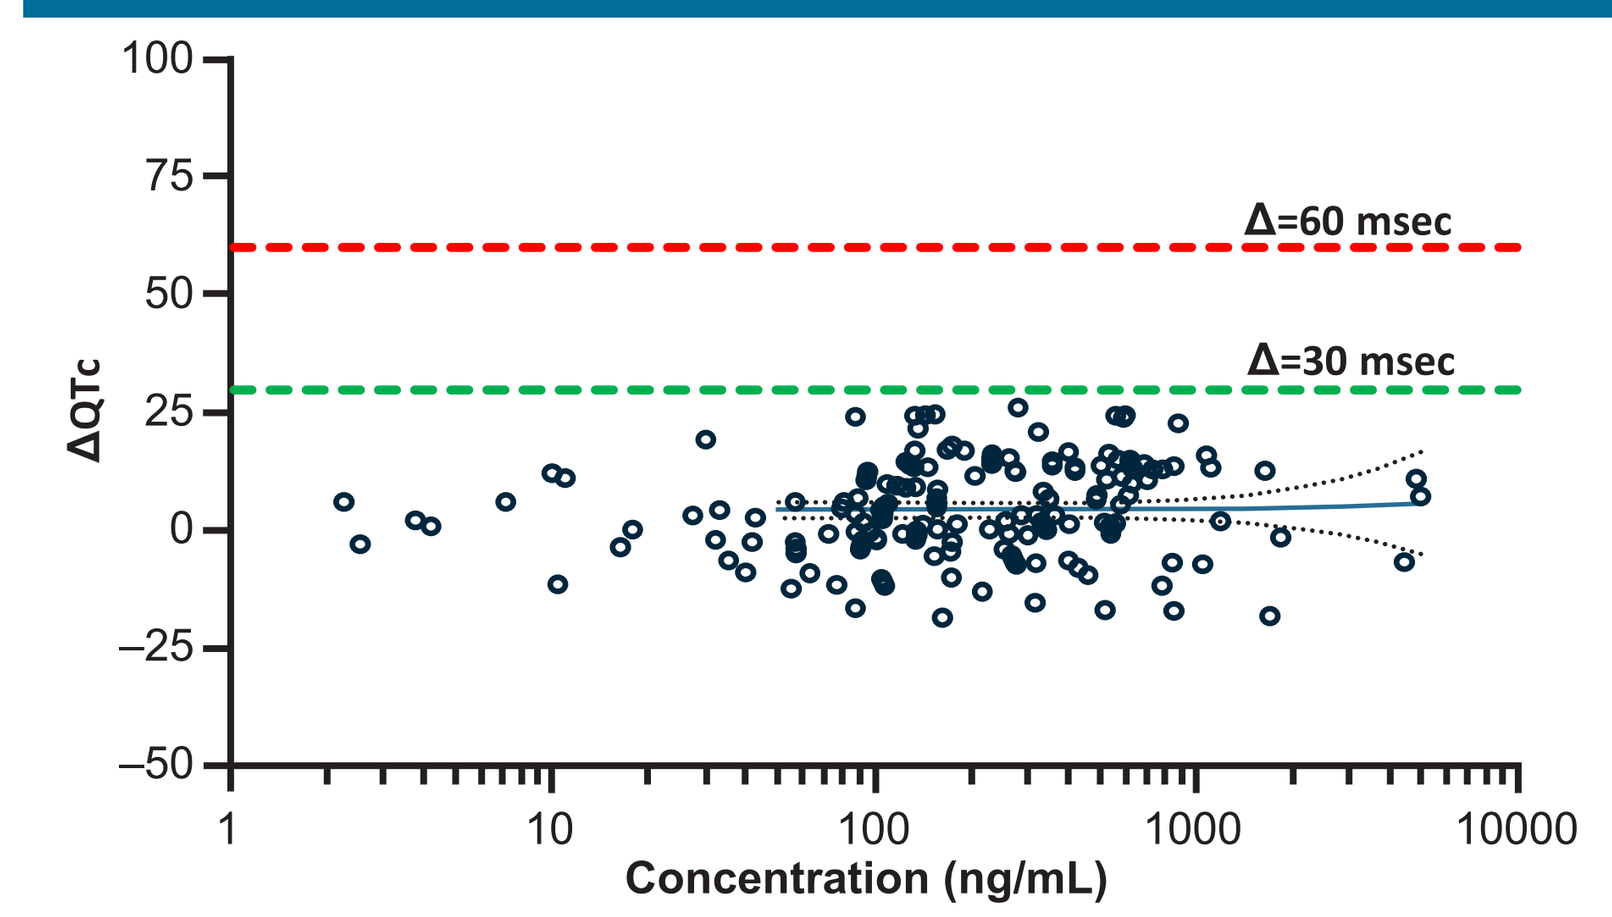

Δ=60 msec
Δ=30 msec
ΔQTc
Concentration (ng/mL)

- Holter data show no exposure–QTc relationship (n=12)
- No patients with 30 msec or 60 msec QTcF increase from baseline

msec, millisecond; QTc, QT corrected for heart rate; QTcF; QT corrected for heart rate using Fridericia's correction; ΔQTc; delta QT corrected for heart rate.

Pharmacokinetics

- Consistent with prior reports, BLU-222 plasma concentrations increased in a dose–proportional trend up to 600 mg BID in monotherapy[4]
- The average effective half–life was 16 hours (calculated from the extent of accumulation, data not shown)
- BLU-222 monotherapy C_{eff} range led to tumor stasis in preclinical OVCAR-3, MKN-1, and T47D models, and correspond to 25%, 25%, and 60% inhibition of pRb S807/811 in these models, respectively (data not shown)

Pharmacodynamics

- Consistent with prior data,[4] in patients treated with BLU-222 monotherapy, dose–proportional reductions in TK1 activity were observed, with highest reductions seen at dose levels of 400 mg or above. Reductions in pRb were also seen in patients treated with BLU-222 at 400 mg and above

Efficacy

- 16 HR+/HER2− breast cancer patients were treated with BLU-222 monotherapy, 7 of whom were treated at doses projected to be in the active range (≥400 mg BID). Of these 7 patients, 3 had evidence of clinical benefit: 2 patients with non–measurable disease remained on study for more than 6 months (229 days and 183 days, ongoing), 1 with significant reduction in disease–related bone pain, and 1 patient with measurable disease had a confirmed partial response (PR)

References

1. Turner et al. *J Clin Oncol.* 2019;37(14):1169–1178; 2. Tadesse S et al. *Drug Discov Today.* 2020;25(2):406–413; 3. Al-Qasem AJ et al. *NPJ Precis Oncol.* 2022;6:68; 4. Patel M et al. Presented at ASCO Annual Meeting 2023; June 2-6, 2023; Chicago, IL. Poster 293. 5. Brown V et al. Presented at AACR Annual Meeting 2022; April 8-13, 2022; New Orleans, LA. Poster 2306; 6. Brown V et al. Presented at SABCS 2022; December 6-10, 2022; San Antonio, TX. Poster P6-10-07; 7. ClinicalTrials.gov. (VELA) Study of BLU-222 in Advanced Solid Tumors. NCT05252416. https://clinicaltrials.gov/ct2/show/NCT05252416. Last update, April 23, 2024; Accessed May 8, 2024; 8. McCartney A et al. *Br J Cancer.* 2020;123:176–177; 9. Malumbres M. *Genome Biol.* 2014;15:122; 10. Samant TS et al. *Clin Pharmacol Ther.* 2018;104(2):374-383. 11. Reichert ZR et al. *Ann Oncol.* 2023; 34(1):111–120; 12. Martinez-Saez O et al. *NPJ Breast Cancer.* 2021;7(1):8; 13. Marshall C et al. *Cancer Res.* 2022;82(22):4107–4113. 14. Moschos M et al. *Drug Des Devel Ther.* 2016;10: 3407–3413; 15. Auso E et al. *Biomedicines.* 2021;9(3):291.

Acknowledgements and Disclosures

Medical writing and editorial support were provided by Srividya Venkitachalam, PhD, ProEd Communications, and was supported by Blueprint Medicines Corporation (BPMC). The research was funded by BPMC. BPMC reviewed and provided feedback on the poster. The authors had full editorial control of the poster and provided their final approval of all content.

https://meetings.asco.org/abstracts-presentations/233574

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

Conclusions

- These data represent the first presentation of the successful combination of a selective CDK2 inhibitor with an approved CDK4/6 inhibitor
- BLU-222 monotherapy was well tolerated in an unselected, heavily pretreated patient population with advanced solid tumors consistent with preclinical data supporting selectivity of BLU-222
- BLU-222 with ribociclib and fulvestrant combination therapy was well tolerated in patients with HR+/HER2− breast cancer, with no DLTs, no BLU-222–related dose discontinuations, only low–grade GI toxicity and low frequency hematologic toxicity
- BLU-222 in combination with ribociclib and fulvestrant resulted in compelling reductions in TK1 and ctDNA, with statistically significant correlation of TK1 reduction with BLU-222 exposure level
- Emerging signs of clinical benefit and antitumor activity in heavily pretreated patients demonstrates potential for BLU-222 in HR+/HER2− breast cancer
- Enrollment is ongoing for dose escalation and optimization of BLU-222 in combination with 400 mg and 600 mg dose levels of ribociclib with fulvestrant. Dose confirmation cohorts for the combination with ribociclib and letrozole are planned

Presented at the American Society of Clinical Oncology (ASCO) Annual Meeting, May 31 – June 4, 2024, Chicago, IL, USA, & Online Please contact medinfo@blueprintmedicines.com for permission to reprint and/or distribute