Overall Survival in Patients with Systemic Mastocytosis with Associated Hematologic Neoplasm Treated with Avapritinib versus Best Available Therapy

Abstract P1013

Andreas Reiter, MD[1]; Jason Gotlib, MD, MS[2]; Iván Álvarez-Twose, MD, PhD[3]; Deepti H. Radia, MD[4]; Johannes Luebke, MD[1]; Priyanka J. Bobbili, ScD, MS[5]; Aolin Wang, PhD[5]; Chelsea Norregaard, MPH[6]; Saša Dimitrijević, PhD[7]; Erin Sullivan, PhD, MPH[6]; Melinda Louie-Gao, PhD[6]; Juliana Schwaab, MD[1]; Ilene A. Galinsky, MSN, ANP-C[8]; Cecelia Perkins, MPH[2]; Wolfgang R. Sperr, MD[9]; Priya Sriskandarajah, MBBS, MRCP, PhD[4]; Andi Chin[5]; Selvam R. Sendhil[5]; Mei Sheng Duh, ScD, MPH[5]; Peter Valent, MD[9]; Daniel J. DeAngelo, MD, PhD[8]

[1]Department of Hematology and Oncology, University Hospital Mannheim, Mannheim, Germany; [2]Stanford Cancer Institute/Stanford University School of Medicine, Stanford, California, United States; [3]Institute of Mastocytosis Studies of Castilla La Mancha (CLMast) — Spanish Reference Center (CSUR) for Mastocytosis and CIBERONC, Virgen del Valle Hospital, Toledo, Spain; [4]Guy's & St Thomas' NHS Foundation Trust, of Guy's Hospital, London, United Kingdom; [5]Analysis Group, Inc., Boston, Massachusetts, United States; [6]Blueprint Medicines Corporation, Cambridge, Massachusetts, United States; [7]Blueprint Medicines Corporation, Zug, Switzerland; [8]Department of Medical Oncology, Dana-Farber Cancer Institute, Boston, Massachusetts, United States; [9]Department of Internal Medicine I, Division of Hematology and Hemostaseology, and Ludwig Boltzmann Institute for Hematology and Oncology, Medical University of Vienna, Vienna, Austria

Background

- Advanced systemic mastocytosis (AdvSM) is a rare myeloid neoplasm commonly characterized by the accumulation of neoplastic mast cells in various organs and tissues[1,2]
- Systemic mastocytosis with an associated hematologic neoplasm (SM-AHN) is the most common subtype of AdvSM diagnosed in approximately 70% of all AdvSM patients[3,4]
- AdvSM patients diagnosed with SM-AHN often have poor prognosis, with a median overall survival (OS) of 2 years[5]
- Avapritinib is a selective KIT D816V inhibitor approved for AdvSM patients in the United States (US)[6] and Europe (for AdvSM patients treated with prior systemic therapy)[7] based on data from two single-arm trials: EXPLORER (Phase 1; NCT02561988) and PATHFINDER (Phase 2; NCT03580655)[8,9]
- No randomized controlled trial (RCT) has been conducted yet to compare the efficacy of avapritinib versus best available therapy (BAT) in patients with SM-AHN

Aim

- The present study (NCT04695431) compared OS between SM-AHN patients treated with avapritinib in the EXPLORER and PATHFINDER single-arm trials versus SM-AHN patients treated with BAT in standard clinical practice

Methods

Data sources

- Clinical trial data (*avapritinib cohort*)
 - Data from patients treated with avapritinib in the safety populations of the EXPLORER and PATHFINDER trials was used (data cut-off: April 20, 2021; data on file, Blueprint Medicines Corporation)
- Real-world data (*BAT cohort*)
 - A global, observational, retrospective chart review study was conducted at 6 study sites (4 European, 2 US) to identify and collect data from SM-AHN patients who received BAT
 - De-identified data from eligible patients were abstracted from patient health records into a standardized electronic case report form from March 26, 2021 to October 4, 2021

Sample selection

- Real-world patients treated with BAT were identified based on the following key inclusion and exclusion criteria, similar to those from EXPLORER and PATHFINDER
- **Inclusion criteria:**
 - Adults (aged ≥18 years) with a diagnosis of AdvSM and documented SM-AHN subtype in their chart
 - Received ≥1 line of systemic therapy (not necessarily as first line) at a participating site on or after January 1, 2009
 - If a patient received multiple lines of therapy at a participating site, data on all available therapies were collected and analyzed
 - The date of initiation of each line of therapy at the participating site was defined as the index date
- **Exclusion criteria:**
 - History of another primary malignancy that was diagnosed or required therapy within 3 years before the index date, except for completely resected basal cell and squamous cell skin cancer, curatively treated localized prostate cancer, and completely resected carcinoma *in situ* in any site
 - Received avapritinib as the first therapy for AdvSM at a participating site

Study endpoint

- OS was defined as the time from avapritinib or BAT initiation to death from any cause
- If alive at the end of the study, patients were censored at the date of last contact (BAT cohort), or at the last known alive date (avapritinib cohort)
- BAT cohort patients treated with avapritinib were censored at avapritinib initiation

Statistical analysis

- Unadjusted OS was assessed using the Kaplan-Meier method
- On-treatment survival rates at specific time points were obtained using the Nelson-Aalen Estimator[10,11]
- Inverse probability of treatment weighting (IPTW) was used to adjust for differences in *a priori* identified key prognostic covariates between treatment cohorts, e.g., age, gender, ECOG score, presence of thrombocytopenia or anaemia at baseline, elevated serum tryptase levels, number and types of prior lines of therapy, among others.
- IPTW-weighted Cox proportional hazards models adjusted for variables that remained unbalanced after weighting and were used to compare OS between the avapritinib and BAT cohorts

Results

Baseline demographics

- This analysis included 119 patients with SM-AHN who were treated with avapritinib and 83 patients treated with BAT (contributing 121 lines of therapy) (**Table 1**)
- Median (range) age was 70.0 (45.0−88.0) for the avapritinib cohort and 70.5 (37.7−87.5) for the BAT cohort
- There were 72 of 119 (60.5%) male patients in the avapritinib cohort versus 63 of 83 (75.9%) in the BAT cohort
- There were 49 (41.2%) lines of therapy contributed from European patients in the avapritinib cohort versus 101 (83.5%) in the BAT cohort; the remaining patients were treated at North American sites

Baseline clinical characteristics

- Mean (SD) ECOG score was 1.3 (0.9) and 1.1 (0.7) for the avapritinib and BAT cohorts, respectively (**Table 1**)
- More patients in the BAT cohort had a diagnosis of thrombocytopenia at baseline (67.8%) compared to the avapritinib cohort (46.2%)
- Among patients who were tested for at least one *S/A/R* mutation, 59.7% of avapritinib patients and 78.8% of BAT patients tested positive for at least one of these mutations

Table 1. Baseline demographics and clinical characteristics

Baseline characteristics, unweighted sample[1]	Avapritinib cohort	BAT cohort	*P* value[2]
Number of unique patients	**N = 119**	**N = 83**	
Number of lines of therapy	**N = 119**	**N = 121**	
Demographic characteristics			
Age (years)			0.519
Mean (SD)	69.3 (8.7)	69.8 (8.2)	--
Median (min, max)	70.0 (45.0, 88.0)	70.5 (37.7, 87.5)	--
Sex, n (%)			
Female	47 (39.5%)	25 (20.7%)	0.002*
Male	72 (60.5%)	96 (79.3%)	0.002*
Region, n (%)			
North America	70 (58.8%)	20 (16.5%)	<0.001*
Europe	49 (41.2%)	101 (83.5%)	<0.001*
Medical history			
Performance status			
ECOG			0.325
n (%)	119 (100.0%)	121 (100.0%)	--
Mean (SD)	1.3 (0.9)	1.1 (0.7)	--
Median (min, max)	1.0 (0.0, 3.0)	1.0 (0.0, 3.0)	--
ECOG category, n (%)			
0	21 (17.6%)	21 (17.4%)	1.000
1	62 (52.1%)	70 (57.9%)	0.444
≥2	36 (30.3%)	30 (24.8%)	0.422
Anemia, n (%)	72 (60.5%)	70 (57.9%)	0.774
Thrombocytopenia, n (%)	55 (46.2%)	82 (67.8%)	0.001*
Disease characteristics			
Skin involvement			
Any skin involvement, n (%)	33 (27.7%)	25 (20.7%)	0.259
Leukocyte count			
≥16 x 10^9/L, n (%)	30 (25.2%)	38 (31.4%)	0.357
Serum tryptase (ng/mL)			
≥125 ng/mL, n (%)	82 (68.9%)	73 (60.3%)	0.210
KIT mutation			
Patients tested, n (%)	115 (96.6%)	82 (98.8%)	0.651
KIT D816V positive, n (%)	109 (94.8%)	82 (100.0%)	0.042*
SRSF2/ASXL1/RUNX1 (S/A/R) mutation panel			
Patients that were tested for at least one mutation, n (%)	119 (100.0%)	66 (79.5%)	<0.001*
Number of mutated genes in S/A/R panel, n (%)			
0	48 (40.3%)	14 (21.2%)	0.013*
1	43 (36.1%)	34 (51.5%)	0.061
≥2	28 (23.5%)	18 (27.3%)	0.699

**P<0.05*

Abbreviations: BAT: best available therapy; ECOG: Eastern Cooperative Oncology Group; max: maximum; min: minimum; S/A/R: SRSF2/ASXL1/RUNX1; SD: standard deviation.

Notes:

[1] The baseline period was defined as 8 weeks leading up to the index date for the avapritinib cohort and the 12 weeks leading up to the index date for the BAT cohort. Descriptive statistics are reported at the line of therapy level for all variables except *KIT* and S/A/R mutations , which are reported at the patient level, since each patient in the BAT cohorts could contribute more than one line of therapy to the analysis.

[2] Comparisons between cohorts were conducted using the Wilcoxon rank-sum test for continuous variables and chi-squared test for categorical variables. For categorical variables with expected counts <5, Fisher's exact tests were used instead of Chi-squared.

Prior systemic therapy

- There were 69 (58.0%) avapritinib patients treated with prior systemic therapy versus 53 (43.8%) patients in the BAT cohort (**Table 2**)
- In the avapritinib cohort, patients were most frequently pretreated with TKIs (51.3%), followed by cytoreductive therapies (19.3%)
 - On an agent-level, avapritinib patients were most frequently pretreated with midostaurin (43.7%) or cladribine (11.8%)
- Patients in the BAT cohort were most frequently pretreated with cytoreductive therapies (28.9%), followed by TKIs (19.8%)
- Agent-level treatment information for the BAT cohort was collected from patients at all study sites except Medizinische Universität Wien in Vienna, Austria (N=9 lines of therapy), where only treatment class information was collected per local regulations
 - Among BAT patients with agent-level information available, patients most frequently received prior treatment with cladribine (18.8%), followed by midostaurin (15.2%)

Table 2. Prior systemic therapy used to treat AdvSM patients

Prior systemic therapy	Avapritinib cohort	BAT cohort	*P* value[1]
Number of unique patients	**N = 119**	**N = 83**	
Number of lines of therapy	**N = 119**	**N = 121**	
Number with prior systemic therapy, n (%)	69 (58.0%)	53 (43.8%)	0.039*
Number of prior lines of systemic therapy received, n (%)			<0.001*
Mean (SD)	0.9 (1.0)	0.1 (0.3)	--
Median (min, max)	1.0 (0.0, 6.0)	0.0 (0.0, 1.0)	--
0	50 (42.0%)	68 (56.2%)	0.039*
1	46 (38.7%)	37 (30.6%)	0.238
2	14 (11.8%)	12 (9.9%)	0.801
≥3	9 (7.6%)	4 (3.3%)	0.165
Prior treatments received, n (%)			
TKI therapy	61 (51.3%)	24 (19.8%)	<0.001*
Cytoreductive therapy	23 (19.3%)	35 (28.9%)	0.113
Biologic therapy	7 (5.9%)	4 (3.3%)	0.373
Agent-level information available[2]	**N = 119**	**N = 112**	--
TKI			
Midostaurin	52 (43.7%)	17 (15.2%)	<0.001*
Dasatinib	2 (1.7%)	7 (6.3%)	0.094
Imatinib	5 (4.2%)	2 (1.8%)	0.447
Ripretinib	3 (2.5%)	1 (0.9%)	0.622
Other[3]	4 (3.4%)	0 (0.0%)	--
Cytoreductive therapy			
Cladribine	14 (11.8%)	21 (18.8%)	0.195
Azacitidine	4 (3.4%)	2 (1.8%)	0.684
Hydroxyurea	8 (6.7%)	6 (5.4%)	0.874
Other[3]	3 (2.5%)	2 (1.7%)	0.682
Biologic therapy			
Interferon-alpha	3 (2.5%)	2 (1.8%)	1.000
Other[3]	2 (1.7%)	1 (0.8%)	0.620

**P<0.05*

Abbreviations: AdvSM: advanced systemic mastocytosis; ECOG: Eastern Cooperative Oncology Group; max: maximum; min: minimum; SD: standard deviation; TKI: tyrosine kinase inhibitor.

Notes:

[1] Comparisons between cohorts were conducted using the Wilcoxon rank-sum test for continuous variables and chi-squared test for categorical variables. For categorical variables with expected counts <5, Fisher's exact tests were used instead of Chi-squared.

[2] The proportion of patients in the BAT cohort was reported among patients from all study sites except Medizinische Universität Wien (Vienna, Austria) (N=9 lines of therapy), where only treatment class information was collected per local regulations. Individual treatments that were observed in ≥2.0% of lines of therapy in any cohort are reported.

[3] Other TKIs included ibrutinib and ruxolitinib. Other cytoreductive therapies included decitabine and chlorambucil. Other biologic therapies included brentuximab vedotin, obinituzumab, pegylated interferon, and rituximab.

Treatment regimens

- In the avapritinib cohort, 50 (42.0%) patients received avapritinib as the first line of therapy
- BAT patients received median (range) of 1 (1−5) lines of therapy at the study site (**Table 3**)
- BAT patients were most frequently treated with TKIs (73 of 121 lines of therapy, 60.3%) or cytoreductive therapies (46 of 121 lines of therapy, 38.0%)
- Among 79 BAT patients with agent-level treatment information available, the most common treatments, overall, were midostaurin (65 of 112 lines of therapy, 58.0%) and cladribine (25 of 112 lines of therapy, 22.3%)
- In 1L, patients were most commonly treated with tyrosine kinase inhibitors; in 2L and 3L+, patients were most commonly treated with cytoreductive agents

Table 3. Summary of treatments received by the BAT cohort

	Overall	1L	2L	3L+
Number of unique patients	**N = 83**	**N = 78**	**N = 31**	**N = 8**
Number of lines of therapy	**N = 121**	**N = 78**	**N = 31**	**N = 12**
Total number of lines of therapy contributed by patient				
Mean (SD)	1.5 (0.8)	1.5 (0.8)	1.3 (0.6)	1.0 (0.0)
Median (min, max)	1.0 (1.0, 5.0)	1.0 (1.0, 5.0)	1.0 (1.0, 2.0)	1.0 (1.0, 1.0)
Number of lines of therapy contributed, n (%)				
1	54 (65.1%)	50 (64.1%)	2 (66.7%)	2 (100.0%)
2	24 (28.9%)	23 (29.5%)	1 (33.3%)	0 (0.0%)
≥3	5 (6.0%)	5 (6.4%)	0 (0.0%)	0 (0.0%)
Year of line of therapy start date, n (%)				
2009–2013	30 (24.8%)	24 (30.8%)	5 (16.1%)	1 (8.3%)
2014–2017	57 (47.1%)	34 (43.6%)	18 (58.1%)	5 (41.7%)
2018–2021	34 (28.1%)	20 (25.6%)	8 (25.8%)	6 (50.0%)
Agents used in each included line of therapy, n (%)				
TKI therapy	73 (60.3%)	54 (69.2%)	15 (48.4%)	4 (33.3%)
Cytoreductive therapy[1]	46 (38.0%)	22 (28.2%)	16 (51.6%)	8 (66.7%)
Biologic therapy[1]	3 (2.5%)	2 (2.6%)	1 (3.2%)	0 (0.0%)
Agent-level information available[2]	**N = 112**	**N = 74**	**N = 28**	**N = 10**
TKI				
Midostaurin	65 (58.0%)	47 (63.5%)	15 (53.6%)	3 (30.0%)
Ripretinib	3 (2.7%)	2 (2.7%)	0 (0.0%)	1 (10.0%)
Ibrutinib	2 (1.8%)	2 (2.7%)	0 (0.0%)	0 (0.0%)
Dasatinib	2 (1.8%)	2 (2.7%)	0 (0.0%)	0 (0.0%)
Cytoreductive therapy				
Cladribine	25 (22.3%)	12 (16.2%)	9 (32.1%)	4 (40.0%)
Hydroxyurea	8 (7.1%)	5 (6.8%)	2 (7.1%)	1 (10.0%)
Azacitidine	3 (2.7%)	0 (0.0%)	2 (7.1%)	1 (10.0%)
Decitabine	2 (1.8%)	2 (2.7%)	0 (0.0%)	0 (0.0%)
Biologic therapy				
Interferon-alfa	1 (0.9%)	1 (1.4%)	0 (0.0%)	0 (0.0%)
Pegylated interferon	1 (0.9%)	0 (0.0%)	1 (3.6%)	0 (0.0%)
Brentuximab vedotin	1 (0.9%)	1 (1.4%)	0 (0.0%)	0 (0.0%)

**P<0.05*

Abbreviations: 1L: first line of therapy; 2L: second line of therapy; 3L+: third or later line of therapy; BAT: best available therapy; max: maximum; min: minimum; SD: standard deviation; TKI: tyrosine kinase inhibitor.

Notes:

[1] In the 2L group, one patient was treated with biologic and cytoreductive agents in same LOT; therefore, the sum of the individual agent class counts (N=32) differs from the number of lines of therapy observed in the 2L sample (N=31). Likewise, in the overall sample, the sum of individual agent class counts (N=122) differs from the total number of lines of therapy observed in the analysis (N=121).

[2] Agent-level information for prior treatments was reported among patients from all study sites except Medical University of Vienna (Austria) (N=9 lines of therapy), where only treatment class information was collected per local regulations.

Overall survival

- During the follow-up period, deaths occurred in 29 (24.4%) avapritinib patients and 56 (67.5%) BAT patients
- In the unweighted analysis, median OS was 46.9 months (95% CI: 44.9, not estimable) in the avapritinib cohort and was 18.0 months (95% CI: 13.0, 26.8) in the BAT cohort (**Figure 1**)
- In the weighted Cox analysis, OS was significantly improved in the avapritinib versus BAT cohort (hazard ratio [HR] [95% CI]: 0.42 [0.24, 0.74]; P<0.001), even with further adjustment for variables that were unbalanced after weighting (**Table 4**)

Figure 1.KM curve for OS among patients with SM-AHN treated with avapritinib versus BAT

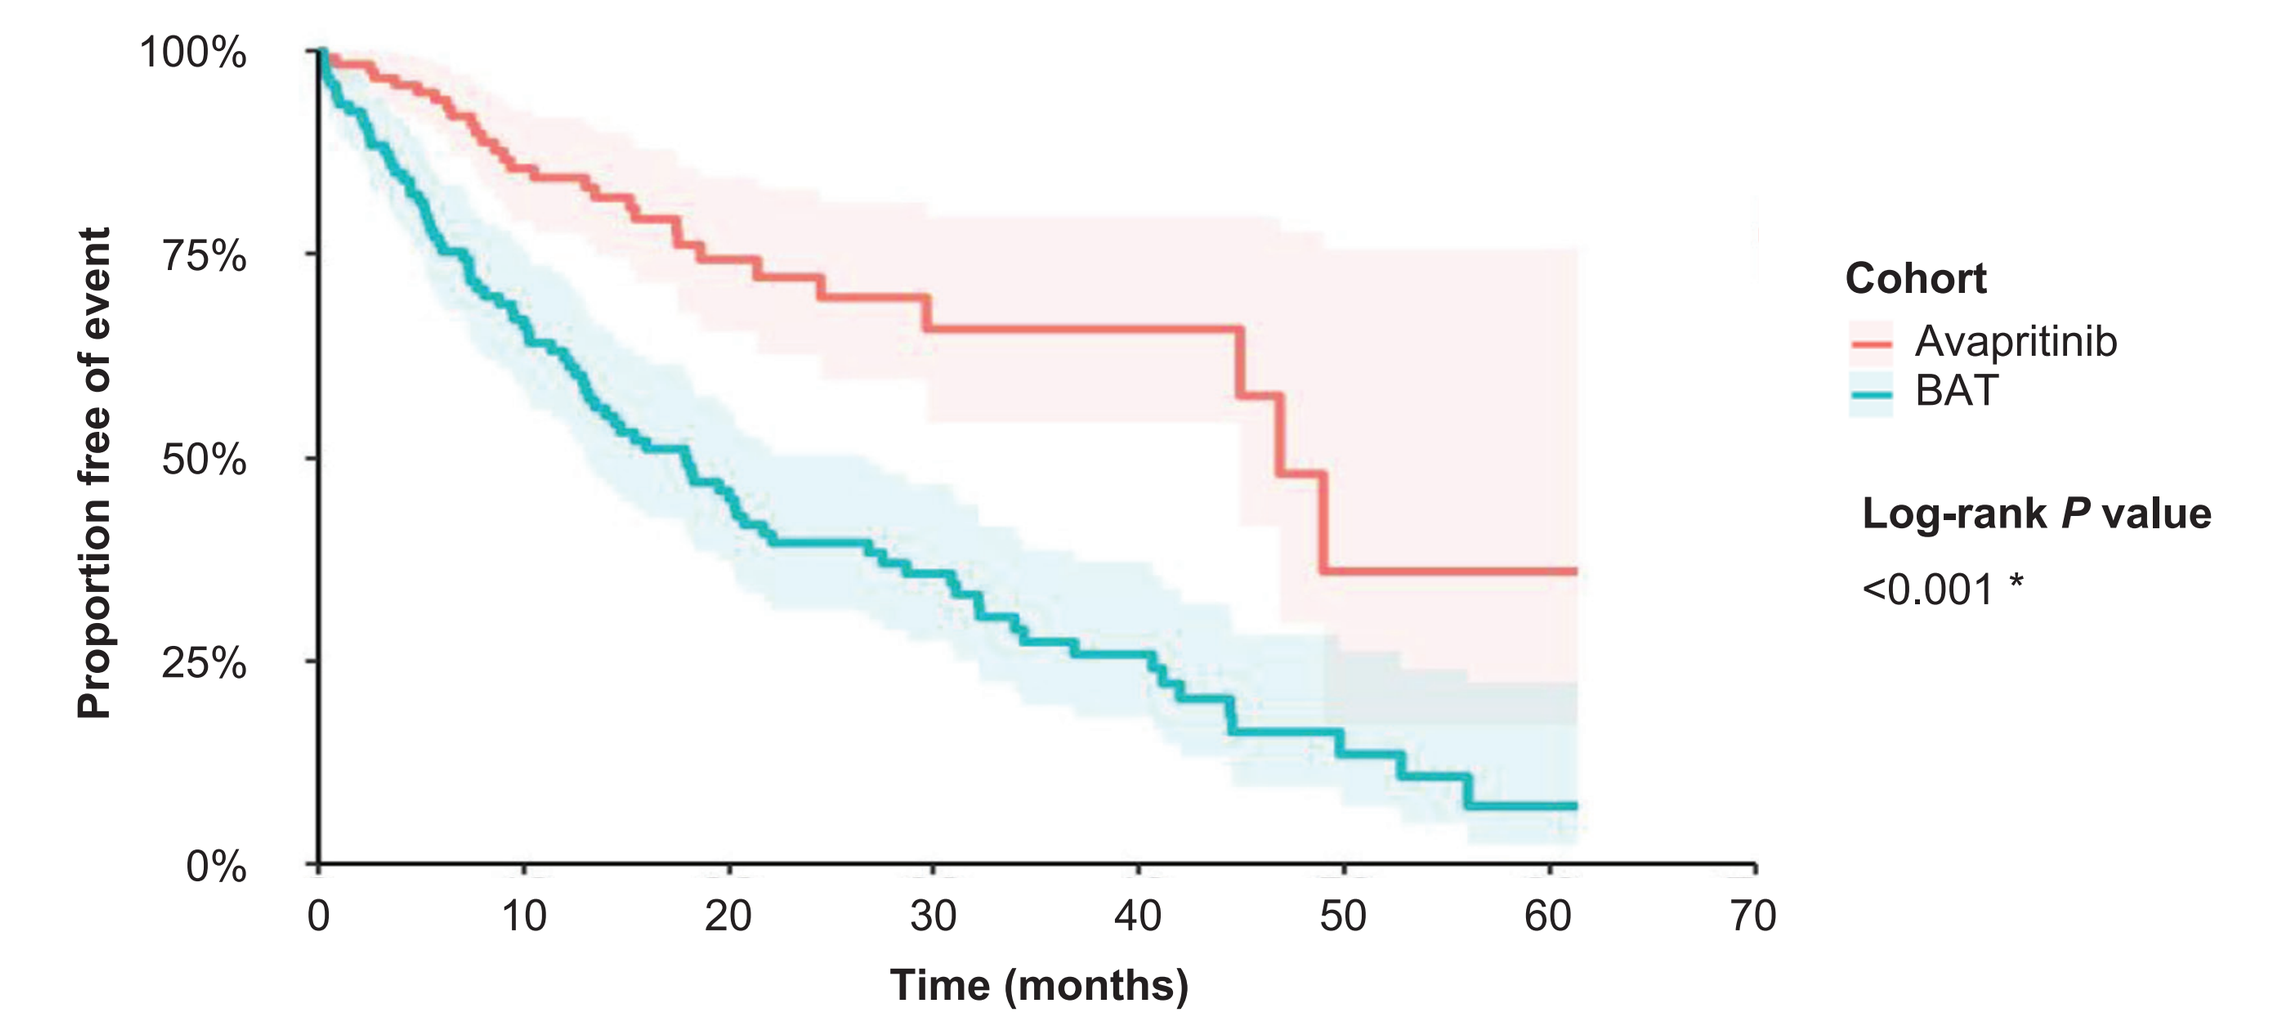
100%
75%
50%
25%
0%
Proportion free of event
0
10
20
30
40
50
60
70
Time (months)
Cohort
Avapritinib
BAT
Log-rank *P* value
<0.001 *

Number at risk	0	10	20	30	40	50	60	70
Avapritinib	119	76	39	16	10	3	1	0
BAT	121	71	44	28	15	5	2	0

**P<0.05*

Abbreviations: BAT: best available therapy; KM: Kaplan-Meier; OS: overall survival; SM-AHN: systemic mastocytosis with an associated hematologic neoplasm.

Note: The follow-up time for the BAT cohort was truncated to match the maximum follow-up time of the avapritinib cohort. In the BAT cohort, 83 patients contributed 121 lines of therapy to the analysis

Table 4. Summary of OS for avapritinib versus BAT

Overall survival	Avapritinib cohort	BAT cohort	*P* value
Number of unique patients	**N = 119**	**N = 83**	
Number of lines of therapy	**N = 119**	**N = 121**	
Deaths from unique patients, n (%)	29 (24.4%)	56 (67.5%)	--
Unique patients censored due to avapritinib initiation, n (%)	--	9 (10.8%)	--
Unique patients censored due to new primary malignancy after index date, n (%)	**--**	**5 (6.0%)**	--
Mean follow-up (months)	17.6	18.1	--
Median OS, unweighted sample (months) (95% CI)	46.9 (44.9, NE)	18.0 (13.0, 26.8)	--
HR, IPTW-weighted sample (95% CI)[1,2]	0.42 (0.24, 0.74)		<0.001 *

**P<0.05*

Abbreviations: CI: confidence interval; ECOG: Eastern Cooperative Oncology Group; HR: hazard ratio; IPTW: inverse probability of treatment weighting; OS: overall survival; NE: not estimable; S/A/R: SRSF2/ASXL1/RUNX1; TKI: tyrosine kinase inhibitor.

Notes:

[1] Stabilized weights were generated using the following baseline characteristics: age, sex, region, ECOG score, anemia (hemoglobin <10 g/dL), thrombocytopenia (platelet count <100 x 10^9/L), skin involvement, leukocyte count ≥16 × 10^9/L, serum tryptase level ≥125 ng/mL, number of mutated genes within the S/A/R panel, number of prior lines of therapy, and prior use of TKI, cytoreductive, or biologic therapy. To reduce variability, stabilized weights were capped at the 1st and 99th percentiles.

[2] The IPTW-weighted Cox proportional hazards model with a robust sandwich variance estimator was used to model overall survival and further adjusted for covariates with a standardized difference >10% after weighting. HR and the corresponding 95% CI and *P* value were presented. Two-sided *P* value <0.05 was considered statistically significant without multiplicity adjustment.

Limitations

- SM-AHN diagnoses for BAT cohort patients were based on local clinician-assessed evaluation; thus, it is possible an incorrect subtype diagnosis was made prior to the substantial increases in disease knowledge that have occurred over the last decade
- SM-AHN diagnoses for trial patients treated with avapritinib were centrally adjudicated, therefore, misclassification of clinician-assessed SM-AHN diagnosis for patients in the BAT cohort may result in an underestimate of differences in OS
 - Since all participating centers hold expertise in AdvSM diagnosis, this concern is mitigated
- Since data collection for BAT cohort patients was conducted retrospectively, results may have been impacted by incomplete reporting for key prognostic characteristics, such as performance status
 - A sensitivity analysis assessing the impact of missing performance status indicated that this is not expected to impact results

Conclusions

- The results of this observational, retrospective study indicate that patients with SM-AHN treated with avapritinib in clinical trials had significantly longer OS compared to patients treated with BAT in standard clinical practice
- SM-AHN is the most common and clinically challenging subtype of AdvSM; in the absence of an RCT, the improved efficacy of avapritinib compared to other systemic treatments observed in this study further validates avapritinib as a treatment option for this patient population

Acknowledgements

This study was funded by Blueprint Medicines Corporation.

References

1. Valent P. Mastocytosis: A paradigmatic example of a rare disease with complex biology and pathology. *Am J Cancer Res.* 2013;3(2):159-172.
2. Ustun C, Arock M, Kluin-Nelemans HC, et al. Advanced systemic mastocytosis: From molecular and genetic progress to clinical practice. *Haematologica*. 2016;101(10):1133-1143.
3. Jawhar M, Dohner K, Kreil S, et al. KIT D816 mutated/CBF-negative acute myeloid leukemia: A poor-risk subtype associated with systemic mastocytosis. *Leukemia*. 2019;33(5):1124-1134.
4. Jawhar M, Schwaab J, Álvarez-Twose I, et al. MARS: Mutation-Adjusted Risk Score for Advanced Systemic Mastocytosis. *Journal of Clinical Oncology.* 2019;37(31):2846-2856.
5. Lim K-H, Tefferi A, Lasho TL, et al. Systemic mastocytosis in 342 consecutive adults: Survival studies and prognostic factors. *Blood*. 2009;113(23):5727-5736.
6. U.S. Food and Drug Administration. FDA approves avapritinib for advanced systemic mastocytosis. In:2021. https://www.fda.gov/drugs/resources-information-approved-drugs/fda-approves-avapritinib-advanced-systemic-mastocytosis
7. Blueprint Medicines Corporation. Blueprint Medicines' AYVAKYT® (avapritinib) Receives European Commission Approval for the Treatment of Adults with Advanced Systemic Mastocytosis. In:2022. https://ir.blueprintmedicines.com/news-releases/news-release-details/blueprint-medicines-ayvakytr-avapritinib-receives-european?utm_source=LinkedIn&utm_medium=social&utm_content=Image&utm_campaign=Corporate_data_milestones
8. DeAngelo DJ, Radia DH, George TI, et al. Safety and efficacy of avapritinib in advanced systemic mastocytosis: The phase 1 EXPLORER trial. *Nat Med.* 2021;27:2183-2191.
9. Gotlib J, Reiter A, Radia DH, et al. Efficacy and safety of avapritinib in advanced systemic mastocytosis: Interim analysis of the phase 2 PATHFINDER trial. *Nat Med.* 2021;27:2192-2199.
10. Nelson W. Hazard plotting for incomplete failure data. *J Qual Technol.* 1969;1(1):27-52.
11. Aalen O. Nonparametric inference for a family of counting processes. *Ann Stat.* 1978;6(4):701-726.
12. Oken MM, Creech RH, Tormey DC, et al. Toxicity and response criteria of the Eastern Cooperative Oncology Group. *Am J Clin Oncol.* 1982;5(6):649-655.

Presented at **EHA 2022 Hybrid Congress**, June 9-12, 2022. Please contact medinfo@blueprintmedicines.com for permission to reprint and/or distribute